Oral semaglutide adherence and associated risk factors in patients with type 2 diabetes in the United States

Lei Lv[1]*; James Amamoo[1]; Yan Wang[2]; Yuanjie Liang[1]; Joanna Boland[2]; Lin Xie[1]; Josh Noone[1]; Mico Guevarra[1]; Yan Song[2]; Alexander Turchin[3]

Aim

- To evaluate adherence to and factors associated with lower adherence to oral semaglutide, with a focus on almost adherent patients with type 2 diabetes (T2D) in a real-world setting.

Introduction

- Among the 38 million adults living with diabetes in the United States (US), an estimated 90–95% have T2D.[1]
 - Glucagon-like peptide-1 (GLP-1) receptor agonists are a well-established treatment option for people with T2D, with oral semaglutide representing the first oral GLP-1 analog approved for treatment of T2D.[2–3]
- Adherence to anti-diabetic medications, including GLP-1 receptor agonists, is integral to their therapeutic effect, with increased adherence associated with a lower incidence of adverse clinical outcomes and increased glycemic control.[4–6] Understanding factors that impact patient adherence to oral semaglutide is essential in forming effective intervention programs in the clinical treatment of T2D.

Methods

- This US-based, retrospective cohort study utilized patient data from Optum's de-identified Clinformatics® Data Mart Database (study period January 2018–June 2023) of adults with T2D who initiated oral semaglutide (full selection criteria shown in **Figure 1**).
- The index date was defined as date of first fill of oral semaglutide on or after September 20, 2019, and patients had continuous follow-up data. The baseline period was defined as the 12-month period prior to the index date, and the follow-up period was defined as the 12-month period after the index date, divided into four quarters (Q).
- Proportion of days covered (PDC) was used to assess adherence (with a stockpiling adjustment) and was evaluated for each Q in the follow-up period.
- Patients were categorized into non-adherent (PDC <48%), almost adherent (48% ≤PDC <80%) and adherent (PDC ≥80%) groups.
 - A cut-off of 48% was chosen, since almost adherence becomes adherence on one additional prescription fill at each follow-up Q: 80% - [(30/91)*100]% = 48% (where 30 days are 1 month of fills, and 91 days = 1 Q).
 - Adherence categories were also assessed in a subgroup analysis of patients aged ≥65 years.
- Multinomial random-effects logistic regression was used to assess risk and protective factors during baseline and follow-up periods of adherence to oral semaglutide.

Results

- A total of 20,016 patients were included in this study; baseline demographic, clinical characteristics and healthcare resource utilization are presented in **Table 1**.
- The prescribing physician specialty was most commonly primary care provider (80.6%), followed by endocrinology (11.8%), other/unknown (7.5%) and cardiology/cardiovascular surgery (0.7%).
- The mean quarterly PDC for Q1–Q4 was 78%, 53%, 46% and 42%, respectively.
- The proportion of adherent and almost adherent patients dropped from Q1 to Q4 (59.8% vs 35.7% and 18.5% vs 7.8%, respectively), while the percentage of non-adherent patients increased (21.8% vs 56.5%; **Figure 2**).
- Factors associated with patients being almost adherent versus adherent are shown in **Figure 3**.
 - Patients of some ethnic minorities and who resided in the South or West were significantly more likely to be almost adherent versus adherent. This was also true for patients with a history of atherosclerotic cardiovascular disease (ASCVD), with HbA_{1c} ≥8% and with anxiety disorder, among other characteristics.

Figure 1: Patient selection

Criterion	n
≥2 diagnoses of T2D (on ≥2 distinct days)	n = 5,638,195
Did not have a diagnosis of T1D at any time	n = 5,128,704
Initiated oral semaglutide after the first observed T2D diagnosis	n = 66,484
Index date on or after September 20, 2019*	n = 66,484
Continuous claims enrollment from ≥12 months both prior to and after the index date	n = 20,036
No diagnosis of gestational diabetes during the baseline or follow-up periods	n = 20,018
Age ≥18 years old on index date	n = 20,016
Date of death is not before the end of the follow-up period†	n = 20,016
	Final sample n = 20,016

*Approval date of oral semaglutide by the US FDA. †Imputed as the 15th of the documented month of death
FDA, Food and Drug Administration; T1D, type 1 diabetes; T2D, type 2 diabetes

Table 1: Baseline demographics, clinical characteristics and HCRU

Characteristic	n (%)*
Male	9,725 (48.6)
Race	
White	11,676 (58.3)
Black	3,072 (15.3)
Hispanic	3,005 (15.0)
Asian	981 (4.9)
Other / unknown	1,282 (6.4)
Geographic region	
South	10,898 (54.4)
West	3,543 (17.7)
Midwest	3,889 (19.4)
Northeast	1,670 (8.3)
Other / unknown	16 (0.1)

Characteristic	n (%)*
Age on index (years)	
≥65	10,553 (52.7)
History of ASCVD	5,623 (28.1)
Obesity	11,149 (55.7)
Evidence of HbA_{1c} ≥8% at baseline or on index	6,301 (51.2)
Anxiety disorder	3,225 (16.1)
Insulin use on index date	2,390 (11.9)
Polypharmacy†	15,786 (78.9)
PDC for any T2D drugs ≥80% during baseline	10,470 (52.3)
GLP-1 RA-naïve before index date	17,152 (85.7)
Number of T2D-related inpatient visits (PPPY), mean (SD)	0.1 (0.4)
Number of non-T2D-related outpatient visits (PPPY), mean (SD)	11.0 (13.9)

*Data displayed as n (%), unless otherwise stated. †Concurrent use of ≥5 medications
ASCVD, atherosclerotic cardiovascular disease; GLP-1 RA, glucagon-like peptide-1 agonist; HbA_{1c}, glycated hemoglobin; HCRU, health-care resource utilization; PDC, proportion of days covered; PPPY, per person per year; SD, standard deviation; T2D, type 2 diabetes

Figure 2: Proportion of adherence, almost adherence and non-adherence of oral semaglutide from Q1 to Q4

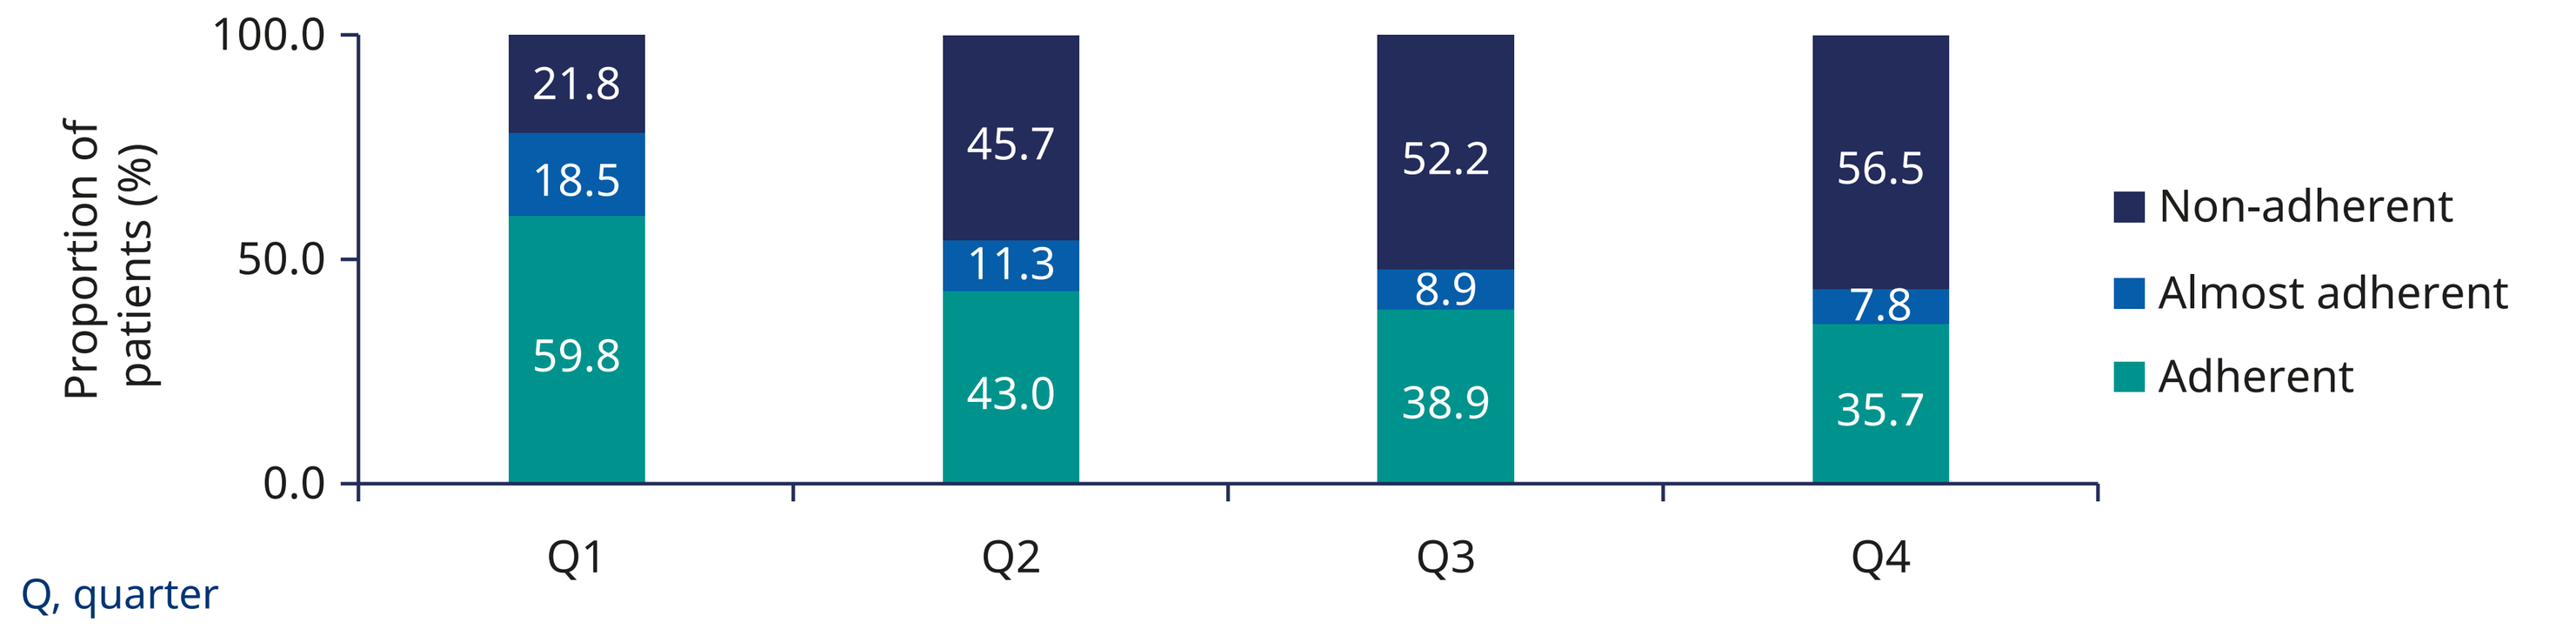
100.0
50.0
0.0
Proportion of patients (%)
21.8
18.5
59.8
45.7
11.3
43.0
52.2
8.9
38.9
56.5
7.8
35.7
Q1
Q2
Q3
Q4
Non-adherent
Almost adherent
Adherent

Q, quarter

- Factors such as polypharmacy, adherence to any T2D drugs, an increased number of non-T2D-related outpatient visits and out-of-pocket costs were significantly associated with a higher likelihood of being adherent.
- Risk factors associated with non-adherence versus adherence were similar to those comparing the almost adherent versus adherent groups, with the addition of older age (65–74 years: OR 1.212, 95% CI 1.085; 1.368, p<0.001; ≥75 years: OR 1.320, 95% CI 1.164; 1.486, p<0.001) acting as a risk factor and obesity acting as a protective factor (OR 0.934, 95% CI 1.875; 1.002, p=0.046).

Figure 3: Risk and protective factors associated with almost adherence versus adherence to oral semaglutide

Key result

Characteristic	OR (95% CI)	p-value for interaction
Baseline characteristics		
Age (reference: <65 years)		
65–74 years	1.084 (0.999; 1.190)	0.060
75+ years	1.094 (0.980; 1.218)	0.090
Sex (reference: male / unknown)		
Female	1.018 (0.950; 1.080)	0.548
Race (reference: White)		
Black	1.240 (1.147; 1.333)	<0.001
Hispanic	1.319 (1.238; 1.416)	<0.001
Asian	1.067 (0.907; 1.224)	0.426
Other/unknown	1.293 (1.153; 1.455)	<0.001
Region (reference: Northeast / other / unknown)		
South	1.419 (1.265; 1.617)	<0.001
West	1.147 (1.016, 1.305)	0.036
Midwest	1.133 (0.988; 1.317)	0.082
Clinical characteristics, medication use and HCRU		
History of ASCVD	1.117 (1.054; 1.190)	<0.001
Obesity	0.999 (0.932; 1.057)	0.978
Evidence of HbA_{1c}≥8%	1.154 (1.081; 1.233)	<0.001
Anxiety disorder	1.101 (1.010; 1.196)	0.028
Insulin use on index date	1.300 (1.164; 1.446)	<0.001
Polypharmacy (excluding ADMs)*	0.792 (0.729; 0.863)	<0.001
PDC for any T2D drugs ≥80%	0.650 (0.611; 0.687)	<0.001
GLP-1 RA-naïve	0.912 (0.823; 1.006)	0.066
Number of T2D-related inpatient visits	0.979 (0.915; 1.059)	0.570
Number of non-T2D-related outpatient visits	0.993 (0.991; 0.996)	<0.001
Receipt of DESC service	0.924 (0.865; 0.990)	0.010
OOP costs, $ (reference: <50)		
50 to <100	0.830 (0.758; 0.904)	<0.001
≥100	0.686 (0.637; 0.742	<0.001

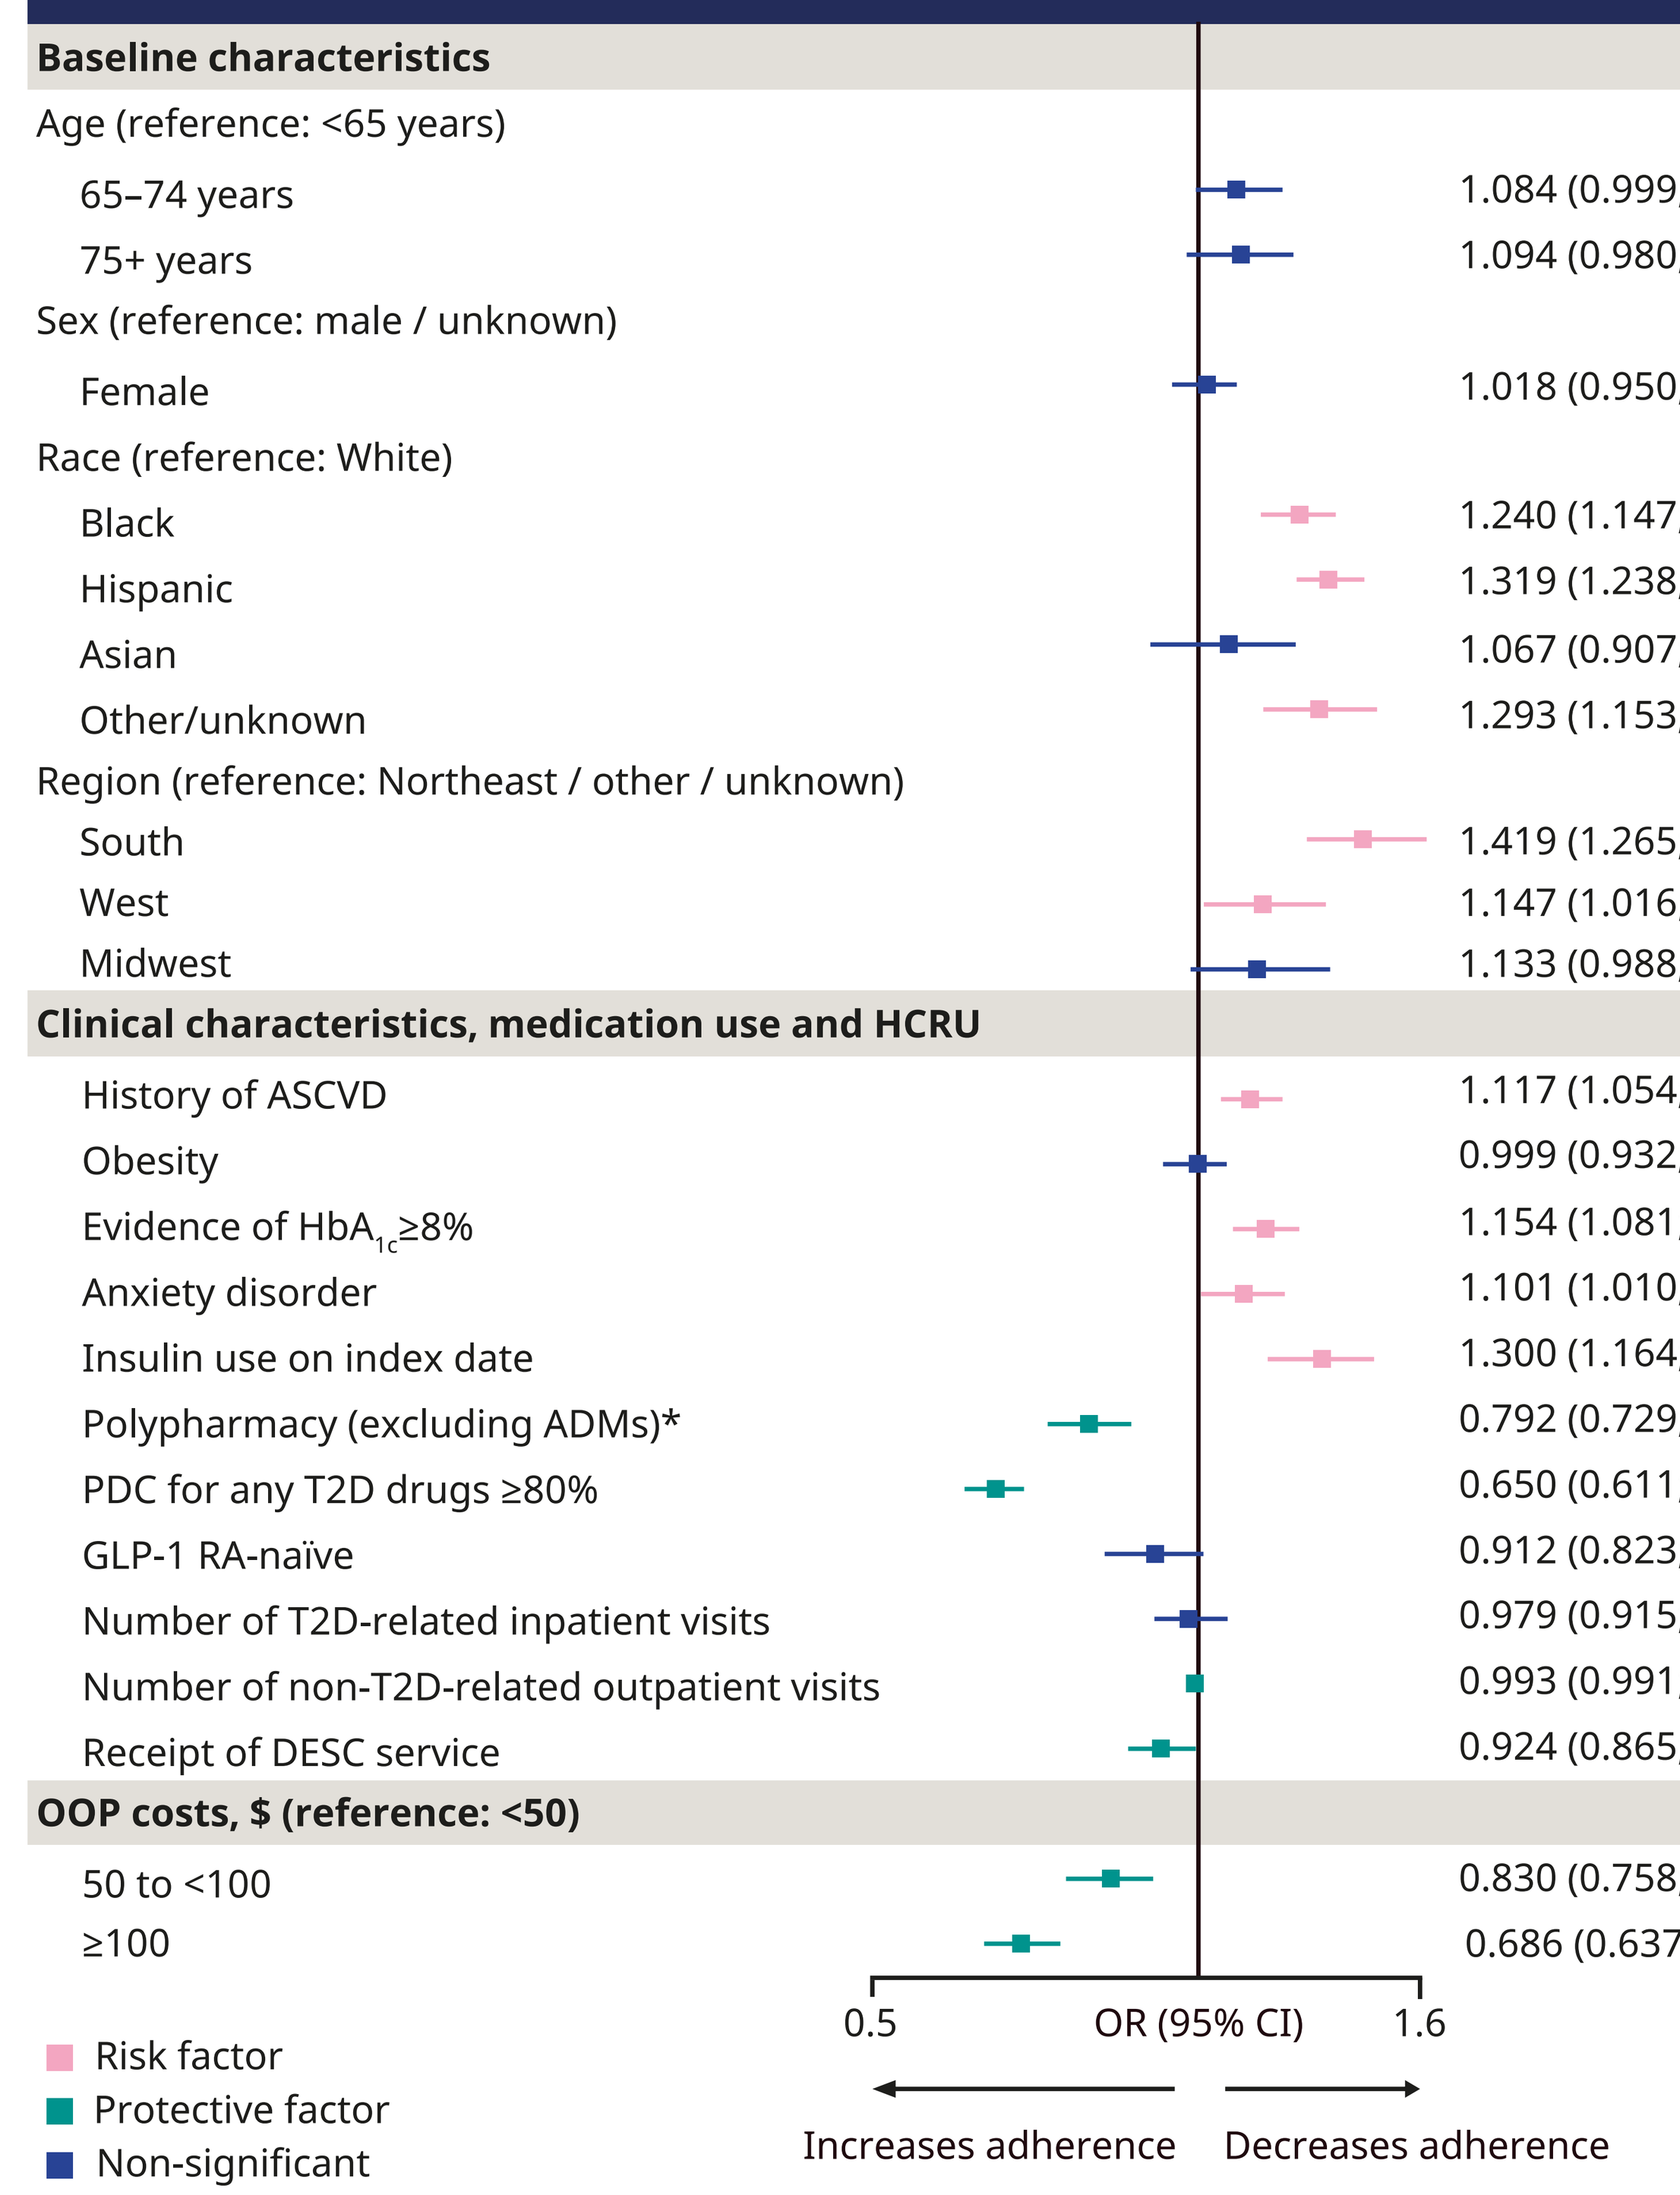
0.5
OR (95% CI)
1.6
Increases adherence
Decreases adherence
Risk factor
Protective factor
Non-significant

*Concurrent use of ≥5 medications
ADM, anti-diabetic medication; ASCVD, atherosclerotic cardiovascular disease; CI, confidence interval; DESC, diabetes education and special care; GLP-1 RA, glucagon-like peptide-1 receptor agonist; HbA_{1c}, glycated hemoglobin; HCRU, healthcare resource utilization; OOP, out-of-pocket; OR, odds ratio; PDC, proportion of days covered; T2D, type 2 diabetes

- In a subgroup analysis of risk and protective factors associated with almost adherence versus adherence in participants ≥65 years (**Figure 4**), risk factors to adherence included history of ASCVD, HbA_{1c} ≥8%, presence of mood disorder and insulin use on index.

Figure 4: Risk and protective factors associated with almost adherence versus adherence to oral semaglutide in participants aged 65 years and over

Characteristic	OR (95% CI)	p-value for interaction
Baseline characteristics		
Age 75+ years (reference: 65–74)	0.959 (0.880; 1.032)	0.352
Female (reference: male / unknown)	1.064 (0.993; 1.133)	0.092
Race (reference: White)		
Black	1.169 (1.049; 1.314)	0.024
Hispanic	1.258 (1.147; 1.374)	<0.001
Asian	1.168 (0.998; 1.390)	0.050
Other/unknown	1.447 (1.269; 1.676)	<0.001
Region (reference: Northeast / other / unknown)		
South	1.613 (1.326; 1.834)	<0.001
West	1.239 (1.110; 1.385)	<0.001
Midwest	1.396 (1.189; 1.580)	<0.001
Clinical characteristics, including medication use		
History of ASCVD	1.106 (1.016; 1.208)	0.026
Obesity	0.946 (0.862; 1.037)	0.248
Evidence of HbA_{1c} ≥8%	1.212 (1.132; 1.303)	<0.001
Anxiety disorder	1.119 (0.989; 1.259)	0.096
Mood disorder	1.390 (1.039; 1.859)	0.042
Number of other ADM classes on index	0.869 (0.830; 0.903)	0.001
Insulin use on index	1.188 (1.056; 1.347)	<0.004
Polypharmacy (excluding ADMs)*	0.814 (0.753; 0.886)	<0.001
PDC for any T2D drugs ≥80%	0.741 (0.688; 0.806)	<0.001
GLP-1 RA-naïve	0.892 (0.788; 1.010)	0.072
OOP costs, $ (reference: <50)		
50 to <100	0.892 (0.764; 1.038)	0.154
≥100	0.612 (0.555; 0.662)	<0.001

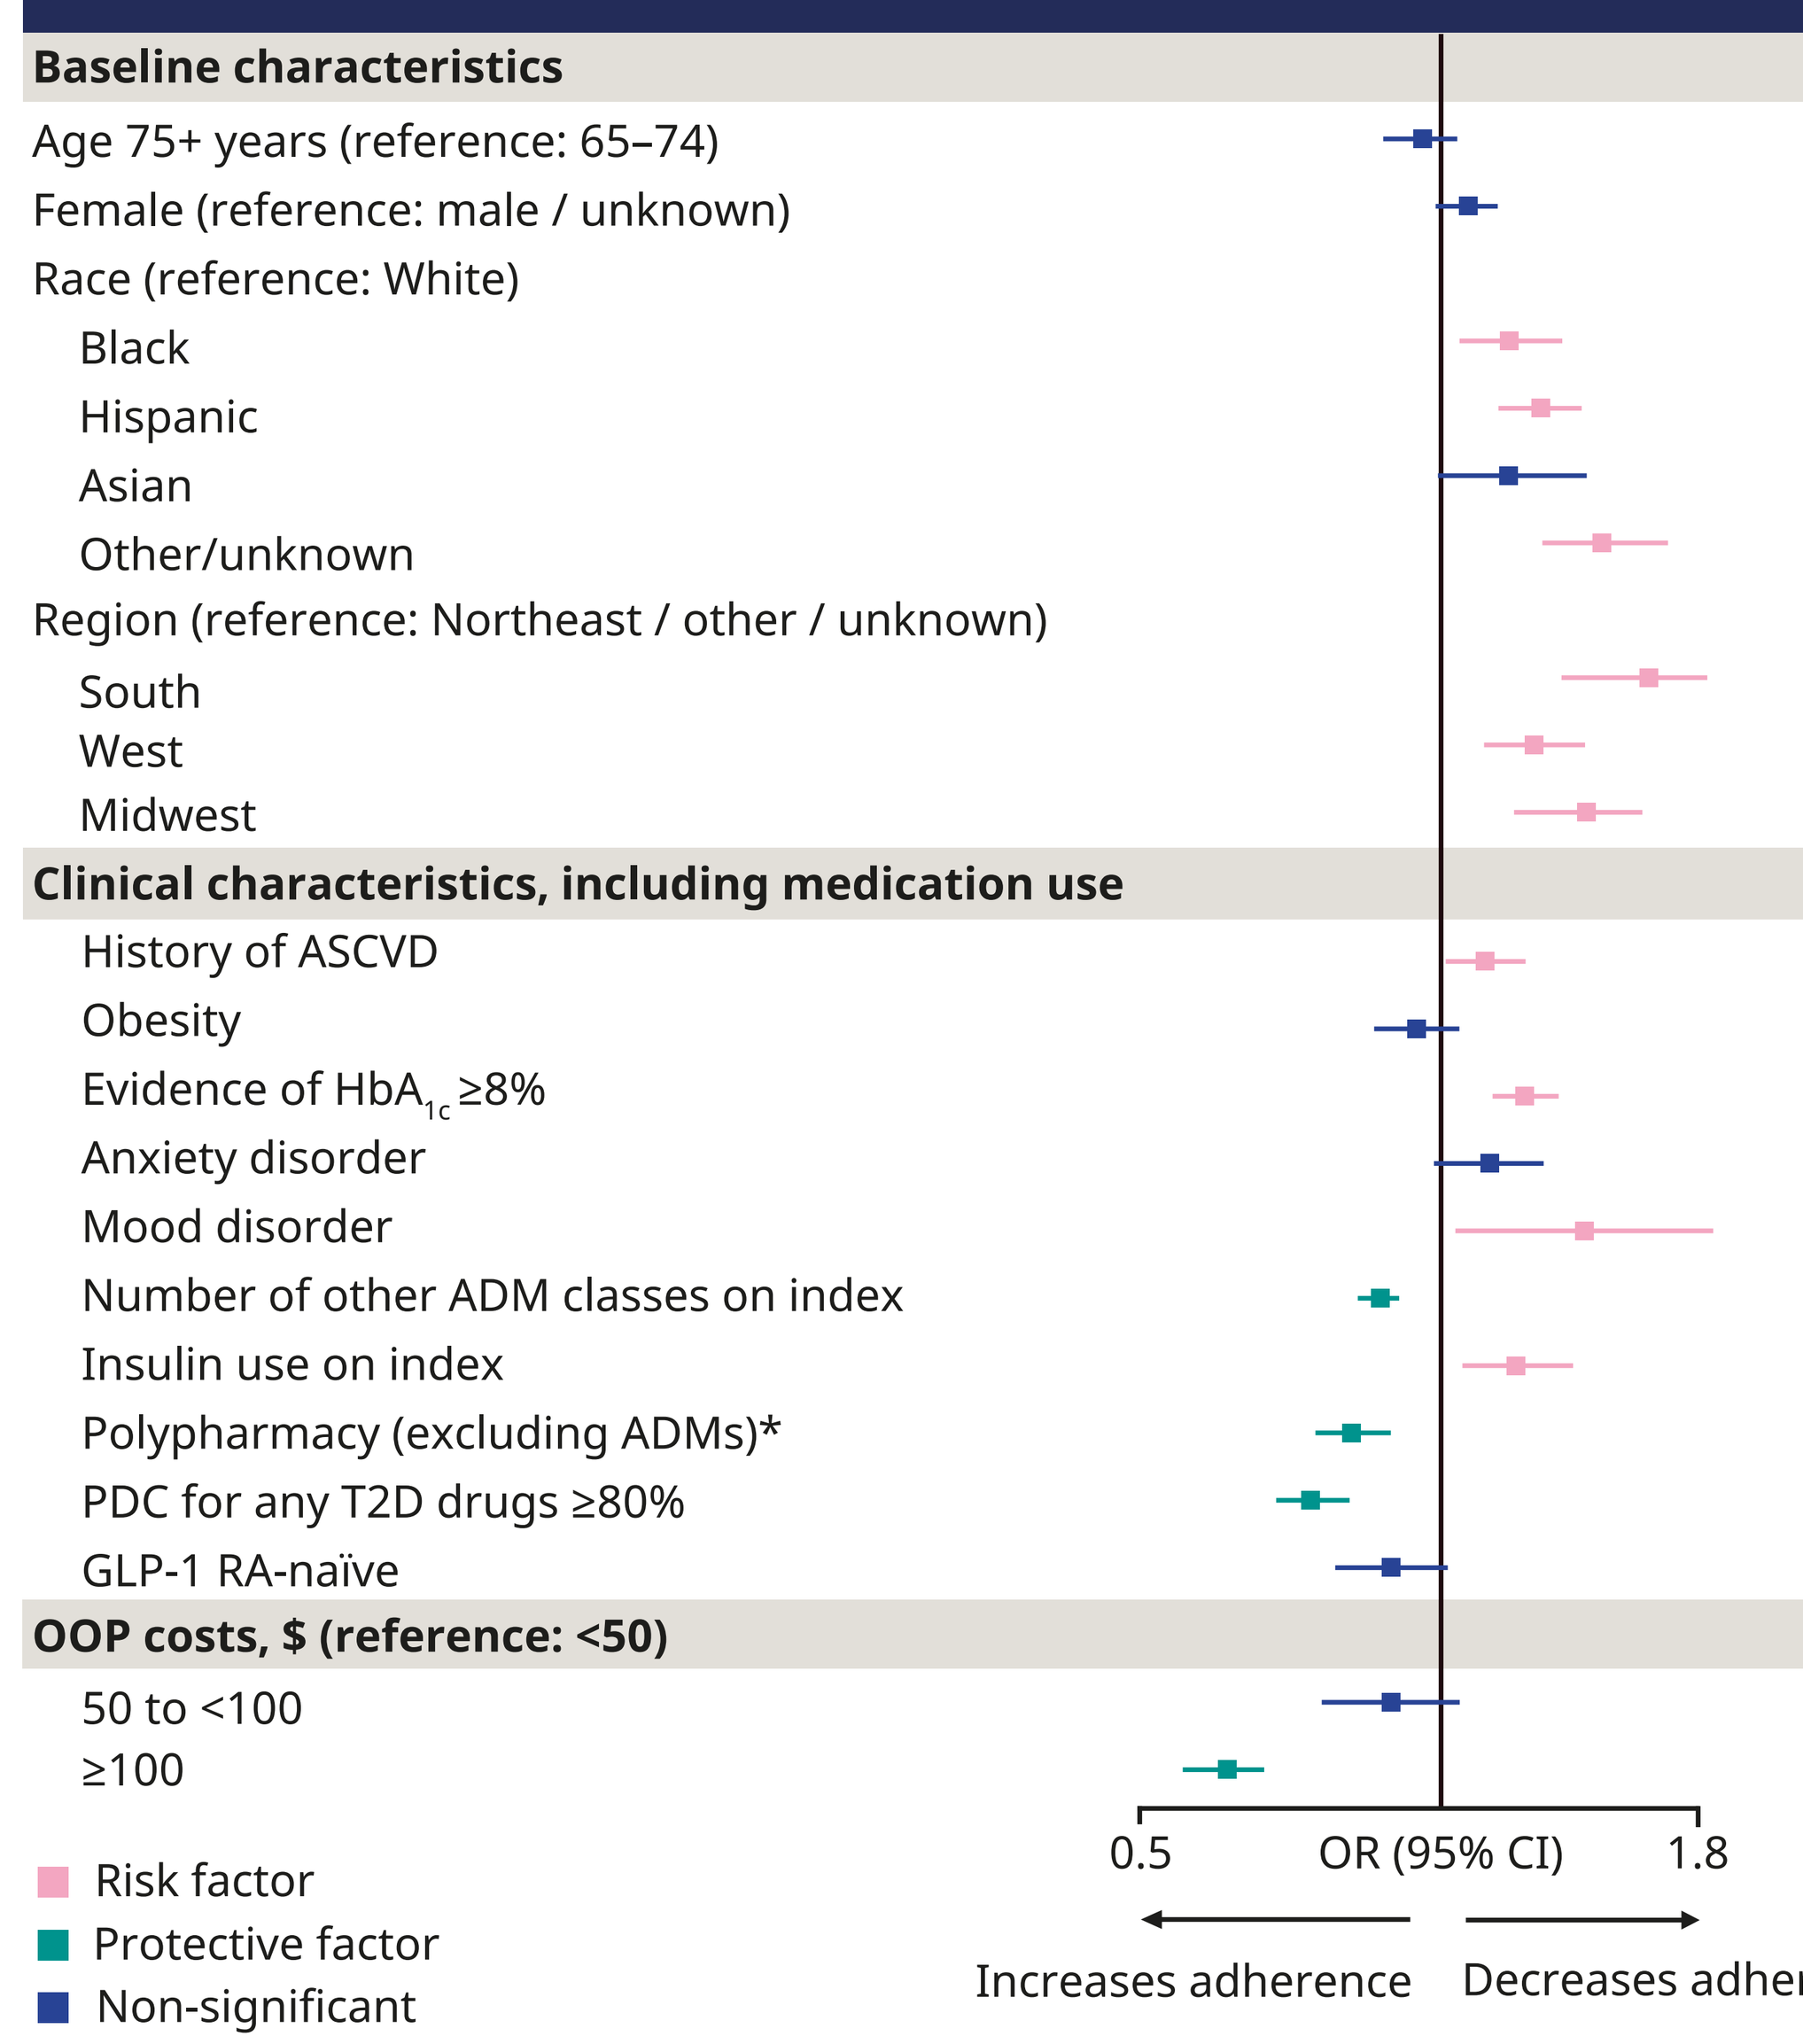
0.5
OR (95% CI)
1.8
Increases adherence
Decreases adherence
Risk factor
Protective factor
Non-significant

*Concurrent use of ≥5 medications
ADM, anti-diabetic medication; ASCVD, atherosclerotic cardiovascular disease; CI, confidence interval; GLP-1 RA, glucagon-like peptide-1 receptor agonist; HbA_{1c}, glycated hemoglobin; OOP, out-of-pocket; OR, odds ratio; PDC, proportion of days covered; T2D, type 2 diabetes

Limitations

- Due to the nature of database studies, the findings reported may include patients who were miscoded and/or misclassified and therefore may not be representative of the overall US population with T2D.

Conclusions

- In improving the identified risk factors associated with adherence to oral semaglutide, these findings may improve patient adherence and form future intervention programs in clinical practice.

[1]Novo Nordisk Inc., Plainsboro, NJ, USA; [2]Analysis Group, Cambridge, MA, USA; [3]Brigham and Women's Hospital, Boston, MA, USA
*Presenting author: Lei Lv, Novo Nordisk Inc., 800 Scudders Mill Road, Plainsboro, NJ 08536,USA. Phone: +1-6098192164; E-mail: lzlv@novonordisk.com
Presented at the Academy of Managed Care Pharmacy (AMCP), March 31–April 03, 2025, Houston, TX, USA

Disclosures:
Lei Lv is an employee of Novo Nordisk Inc.
This study was sponsored by Novo Nordisk Inc. The authors acknowledge the medical writing and editorial assistance of Mhairi Mcgarva, BSc, and Izabel James, MBBS, of Ashfield MedComms, an Inizio company (funded by Novo Nordisk Inc.)

References:
(**1**) US Centers for Disease Control and Prevention. Diabetes Basics. https://www.cdc.gov/diabetes/about/index.html (accessed Jan 9, 2025); (**2**) Isaacs et al. Diabetes Spectr 2021:34;7–19;
(**3**) Novo Nordisk. Rybelsus®. https://www.rybelsus.com/prescribing-information (accessed Jan 9, 2025]; (**4**) Ciardullo et al. Diabetes Metab Res Rev 2024;40:e3791;
(**5**) Palanca et al. Diabetes Ther 2023:14;723–36 (**6**) Giorgino et al. Patient Prefer Adherence 2018:12;707–19