Adherence to once-weekly injectable semaglutide and associated risk factors in patients with type 2 diabetes in the United States

James Amamoo[1]*; Lei Lv;[1] Yan Wang[2]; Yuanjie Liang[1]; Joanna Boland[2]; Lin Xie[1]; Josh Noone[1]; Mico Guevarra[1]; Yan Song[2]; Cory Gamble[1]; Alexander Turchin[3]

Aim

- To investigate adherence to and factors associated with adherence to semaglutide once-weekly (OW) in adults with type 2 diabetes (T2D) in the United States (US), using a large real-world database.

Introduction

- In the US, ~38 million people have diabetes, of whom 90–95% have T2D.[1]
- Semaglutide OW is a glucagon-like peptide-1 receptor agonist (GLP-1 RA) approved for use in adults with 1) T2D to improve glycemic control, as an adjunct to diet and exercise; 2) T2D and established cardiovascular (CV) disease, to reduce the risk of major adverse cardiovascular events; 3) T2D and chronic kidney disease, to reduce the risk of sustained estimated glomerular filtration rate (eGFR) decline, end-stage kidney disease and CV death.[2]
- Adherence to GLP-1 RA therapy is associated with improved glycemic control and a lower incidence of adverse clinical outcomes.[3,4] However, there is limited real-world evidence of the factors associated with adherence to GLP-1 RA therapy in people with T2D.[5,6]

Methods

- This was a retrospective study using administrative claims data from the Optum Clinformatics® Data Mart (study period Jan 1, 2018–Jun 30, 2023) of adults with T2D in the US who initiated semaglutide OW (full selection criteria shown in **Figure 1**).
- The date of the first semaglutide OW pharmacy claim was the index date (on or after Jan 1, 2019); the baseline period was defined as the 12-month period prior to the index date, and the follow-up period was defined as the 12-month period from the index date and was divided into four quarters (Q).
- Adherence to semaglutide OW was assessed using modified proportion of days covered (mPDC), with a stockpiling and titration adjustment (i.e. if the patient's first claim was 0.5 mg, we assume the patient started the treatment with 0.25 mg).
 - mPDC was evaluated separately for each Q in the follow-up period.
- Patients were categorized into three adherence levels: non-adherent (mPDC <50%), almost adherent (50% ≤mPDC <80%) and adherent (mPDC ≥80%).
 - A cut-off of 50% was chosen, since almost adherent becomes adherent at one additional fill at each follow-up Q: 80% - ([28/91]*100)% = 50% (where 28 days are 4 weeks of fills, and 91 days = 1 Q).
- A mixed-effects multivariable multinomial logistic regression model was used to assess the associations of factors with adherence to semaglutide OW in each Q during the baseline or follow-up periods.
 - Factors could either be protective of adherence or risk adherence, i.e. associated with patients being more or less likely to be adherent to semaglutide OW treatment.
 - A subgroup analysis of patients aged ≥65 years was also carried out.

Results

- The final cohort consisted of 70,693 patients; baseline demographics and clinical characteristics are presented in **Table 1**.
- The prescribing physician specialty was most commonly primary care provider (72.1%), followed by endocrinology (20.8%), other/unknown (6.9%) and cardiology/cardiovascular surgery (1.1%).
- The mean quarterly mPDC was 86%, 63%, 57% and 53% for Q1–Q4, respectively.
 - Percentages of both adherent and almost adherent patients dropped from Q1 to Q4 (**Figure 2**).
- **Figure 3** shows factors associated with patients being more or less likely to be almost adherent versus adherent.
 - Risk factors for patients being significantly more likely to be almost adherent than adherent included age, some minority ethnicities and living outside the Northeast, as well as several clinical characteristics and number of T2D-related inpatient visits.

Figure 1: Patient selection

Criterion	n
≥2 diagnoses of T2D on ≥2 distinct days	n = 5,638,195
Did not have a diagnosis of T1D at any time	n = 5,128,704
Initiated semaglutide OW after the first observed T2D diagnosis	n = 267,421
Index date on or after January 1, 2019	n = 266,119
Continuous claims enrollment from ≥12 months both prior to and after the index date	n = 70,796
Did not have a general diagnosis of gestational diabetes during the baseline or follow-up periods	n = 70,698
Date of death is not before the end of the follow-up period	n = 70,693
Age ≥18 years old on index date	n = 70,693
Final sample	n = 70,693

OW, once-weekly; T1D, type 1 diabetes; T2D, type 2 diabetes

Table 1: Baseline demographics, clinical characteristics and HCRU

Characteristic	n (%)*
Male	32,069 (45.4)
Race	
White	45,713 (64.7)
Black	10,324 (14.6)
Hispanic	8,986 (12.7)
Asian	1,819 (2.6)
Other / unknown	3,851 (5.4)
Geographic region	
South	39,291 (55.6)
West	10,783 (15.3)
Midwest	14,725 (20.8)
Northeast	5,869 (8.3)
Other / unknown	25 (0.0)

Characteristic	n (%)*
Age on index (years)	
≥65	33,120 (46.9)
Year of index	
2019	12,611 (17.8)
2020	16,060 (22.7)
2021	24,074 (34.1)
2022	17,948 (25.4)
Insulin use on index date	17,797 (25.2)
Polypharmacy (excluding ADMs)†	38,566 (54.6)
PDC for any T2D drugs ≥80% during baseline	37,458 (53.0)
Number of T2D-related inpatient visits (PPPY), mean (SD)	0.1 (0.5)
Number of non-T2D-related outpatient visits (PPPY), mean (SD)	12.8 (5.4)

*Unless otherwise stated. †Concurrent use of ≥5 medications
ADM, anti-diabetic medication; HCRU, healthcare resource utilization; PDC, proportion of days covered; PPPY, per patient per year; SD, standard deviation; T2D, type 2 diabetes

Figure 2: Adherence categories from Q1 to Q4

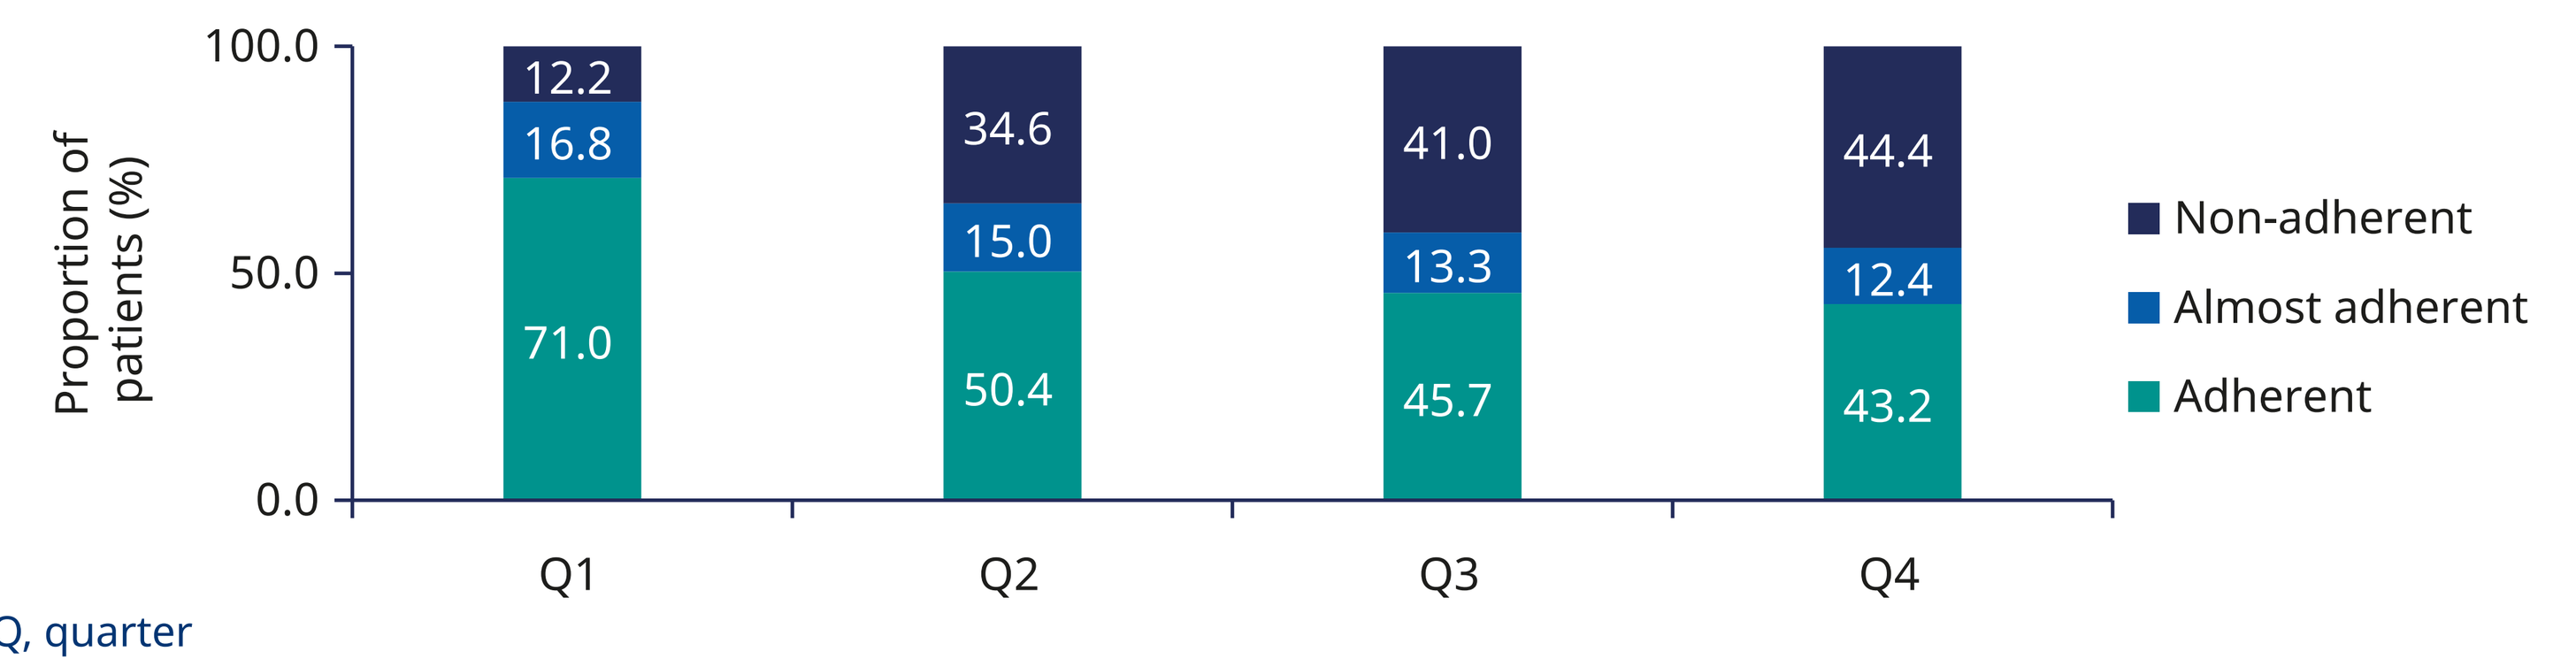
100.0
50.0
0.0
Proportion of patients (%)
12.2
16.8
71.0
34.6
15.0
50.4
41.0
13.3
45.7
44.4
12.4
43.2
Q1
Q2
Q3
Q4
Non-adherent
Almost adherent
Adherent

Q, quarter

- Protective factors were index year (2020 or 2021), polypharmacy, proportion of days covered (PDC) ≥80% for any T2D drugs, obesity and the number of non-T2D-related outpatient visits.
- For non-adherent versus adherent to semaglutide OW, clinical characteristics that were risk factors (OR [95% CI]) included a history of atherosclerotic CV disease (1.136 [1.085; 1.185], p<0.001), evidence of HbA_{1c} ≥8% (1.283 [1.225; 1.344], p<0.001), anxiety disorder (1.076 [1.020; 1.131], p=0.002), insulin use on index (1.116 [1.066; 1.170], p<0.001) and GLP-1 RA-naïve (1.597 [1.491; 1.697], p<0.001).

Figure 3: Risk and protective factors for almost adherent versus adherent to semaglutide OW

Key result

Characteristic	OR (95% CI)	p-value for interaction
Baseline characteristics		
Age, years (reference: <65)		
65–74	1.109 (1.061; 1.156)	<0.001
75+	1.240 (1.167; 1.314)	<0.001
Race (reference: White)		
Black	1.233 (1.185; 1.285)	<0.001
Hispanic	1.209 (1.148; 1.269)	<0.001
Asian	0.983 (0.899; 1.082)	0.694
Other / unknown	1.263 (1.178; 1.335)	<0.001
Region (reference: Northeast / other / unknown)		
South	1.232 (1.156; 1.314)	<0.001
West	1.154 (1.086; 1.226)	<0.001
Midwest	1.136 (1.066; 1.211)	<0.001
Year of index (reference: 2022)		
2019	1.069 (1.007; 1.131)	0.020
2020	0.863 (0.827; 0.901)	<0.001
2021	0.900 (0.860; 0.944)	<0.001
Clinical characteristics, medications and HCRU		
History of ASCVD	1.057 (1.024; 1.108)	<0.001
Obesity	0.950 (0.921; 0.982)	0.004
Evidence of HbA_{1c} ≥8%	1.174 (1.129; 1.217)	<0.001
Anxiety disorder	1.056 (1.021; 1.099)	<0.001
Insulin use on index date	1.104 (1.063; 1.154)	<0.001
Polypharmacy (excluding ADMs)*	0.798 (0.772; 0.833)	<0.001
PDC for any T2D drugs ≥80%	0.686 (0.659; 0.709)	<0.001
GLP-1 RA-naïve on index	1.201 (1.164; 1.239)	<0.001
Number of T2D-related inpatient visits	1.049 (1.015; 1.089)	0.004
Number of non-T2D-related outpatient visits	0.995 (0.994; 0.996)	<0.001
OOP costs, $ (reference: <50)		
50 to <100	1.063 (1.017; 1.112)	0.016
≥100	0.796 (0.769; 0.823)	<0.001

Forest plot axis: 0.6 – OR (95% CI) – 1.4; ← Increased adherence | Decreased adherence →
Risk factor; Protective factor; Non-significant

*Concurrent use of ≥5 medications
ADM, anti-diabetic medication; ASCVD, atherosclerotic cardiovascular disease; CI, confidence interval; GLP-1 RA, glucagon-like peptide-1 receptor agonist; HbA_{1c}, glycated hemoglobin; HCRU, healthcare resource utilization; OOP, out-of-pocket; OR, odds ratio; OW, once-weekly; PDC, proportion of days covered; T2D, type 2 diabetes

- In a subgroup of patients aged ≥65 years (**Figure 4**), risk factors for almost adherent versus adherent included evidence of HbA_{1c} ≥8%, insulin use on index and GLP-1 RA-naïve, whereas protective factors included obesity, number of anti-diabetic medication classes on index, polypharmacy, PDC for any T2D drugs ≥80% and out-of-pocket costs ≥$100.

Figure 4: Risk and protective factors for almost adherent versus adherent to semaglutide OW among a subgroup of patients aged ≥65 years

Characteristic	OR (95% CI)	p-value for interaction
Baseline characteristics		
Age 75+ years (reference: 65–74)	1.110 (1.047; 1.186)	<0.001
Female (reference: male/unknown)	1.020 (0.979; 1.073)	0.460
Race (reference: White)		
Black	1.175 (1.096; 1.252)	<0.001
Hispanic	1.259 (1.187; 1.331)	<0.001
Asian	0.970 (0.851; 1.107)	0.654
Other / unknown	1.239 (1.127; 1.352)	<0.001
Region (reference: Northeast / other / unknown)		
South	1.314 (1.227; 1.400)	<0.001
West	1.194 (1.103; 1.290)	<0.001
Midwest	1.172 (1.061; 1.272)	<0.001
Clinical characteristics, including medication use		
History of ASCVD	1.057 (0.998; 1.108)	0.064
Obesity	0.930 (0.894; 0.973)	0.002
Evidence of HbA_{1c} ≥8%	1.230 (1.171; 1.297)	<0.001
Anxiety disorder	1.035 (0.969; 1.098)	0.266
Mood disorder	1.228 (1.007; 1.462)	0.050
Number of other ADM classes on index	0.908 (0.888; 0.936)	<0.001
Insulin use on index	1.096 (1.038; 1.148)	<0.001
Polypharmacy (excluding ADMs)*	0.809 (0.772; 0.848)	<0.001
PDC for any T2D drugs	0.771 (0.720; 0.821)	<0.001
GLP-1 RA-naïve ≥80%	1.348 (1.283; 1.413)	<0.001
OOP costs, $ (reference: <50)		
50 to <100	1.004 (0.947; 1.065)	0.870
≥100	0.769 (0.730; 0.806)	<0.001

Forest plot axis: 0.7 – OR (95% CI) – 1.5; ← Increased adherence | Decreased adherence →
Risk factor; Protective factor; Non-significant

*Concurrent use of ≥5 medications
ADM, anti-diabetic medication; ASCVD, atherosclerotic cardiovascular disease; CI, confidence interval; GLP-1 RA, glucagon-like peptide-1 receptor agonist; HbA_{1c}, glycated hemoglobin; OOP, out-of-pocket; OR, odds ratio; OW, once-weekly; PDC, proportion of days covered; T2D, type 2 diabetes

Limitations

- Study limitations related to the study design and data source utilized include the potential for miscoding/misclassification and that the findings may not be representative of the overall US T2D population due to insurance type.

Conclusions

- Multiple factors are associated with risking adherence to semaglutide OW treatment, but addressing several of the modifiable factors may help more patients to be adherent, and as such, could be used to formulate interventions to improve adherence.

[1]Novo Nordisk Inc., Plainsboro, NJ, USA; [2]Analysis Group, Cambridge, MA, USA; [3]Brigham and Women's Hospital, Boston, MA, USA
*Presenting author: James Amamoo, Novo Nordisk Inc., 800 Scudders Mill Rd, Plainsboro, New Jersey 08536, USA. Phone: +1-6094809988; Email: jmjw@novonordisk.com
Presented at the Academy of Managed Care Pharmacy (AMCP), March 31–April 03, 2025, Houston, TX, USA

Disclosures:
James Amamoo is an employee of Novo Nordisk. This study was sponsored by Novo Nordisk Inc.
The authors acknowledge the medical writing and editing assistance of Aislinn Harkin, MSci, and Izabel James, MBBS, of Ashfield MedComms, an Inizio company (funded by Novo Nordisk Inc.

References:
(**1**) US Centers for Disease Control and Prevention. Diabetes Basics. https://www.cdc.gov/diabetes/about/about-type-2-diabetes.html (accessed Jan 14, 2025); (**2**) Novo Nordisk. Ozempic® prescribing information. https://www.novo-pi.com/ozempic.pdf (accessed Feb 10, 2025); (**3**) Ciardullo et al. Diabetes Metab Res Rev 2024;40:e3791; (**4**) Palanca et al. Diabetes Ther 2023;14:723–36; (**5**) Weiss et al. Patient Prefer Adherence 2020;14:2337–45; (**6**) Weiss et al. BMJ Open Diabetes Res Care 2022;10:e002517