Healthcare Resource Utilization and Costs in Patients with Metabolic Dysfunction-Associated Steatohepatitis: A Comparison Between Patients with Probable MASH, Without MASH, and Diagnosed MASH

Caichen Zhong[1]; Nana Boame[2]; Semiu O. Gbadamosi[1]; Maximilian Jara[3]; Rakesh Luthra[4]; Alex Salamun[2]; Bret Zeldow[2]; Abdalla Aly[1]; Mary E. Rinella[5]

[1]Real-World Evidence, Clinical Data Science and Evidence, Novo Nordisk Inc., Plainsboro, NJ; [2]Genesis Research Group, Hoboken, NJ; [3]Medical Affairs, Novo Nordisk Inc., Plainsboro, NJ; [4]HEOR Evidence Strategy and Synthesis, Novo Nordisk Inc., Plainsboro, NJ; [5]University of Chicago Pritzker School of Medicine, Chicago, IL.

Aim

To compare healthcare resource utilization (HCRU) and costs among patients with probable MASH, diagnosed MASH, and those without MASH.

Introduction

- Metabolic dysfunction-associated steatohepatitis (MASH) is associated with a high economic burden.[1]
- Most studies have reported on the HCRU and costs in patients with diagnosed MASH.
- However, MASH is often underdiagnosed, with many patients being diagnosed with more advanced disease and often with the presentation of associated comorbidities.[2]
- There is a critical unmet need to characterize the economic burden in patients with undiagnosed MASH, hereafter referred to as probable MASH.

Methods

Study design and data source

- This was a retrospective observational real-world study using claims data from the Komodo Healthcare Map™ (Jan 2016 – Dec 2023). Patients were identified from January 1, 2017 to December 31, 2022.
- Index date was defined per cohort: 1) patients with diagnosed MASH: date of first MASH diagnosis; 2) patients with probable MASH and 3) patients without MASH: date of first qualifying alanine aminotransferase (ALT) lab measurement.
- The baseline period included the 12 months preceding the index date, and the follow-up period was ≥12 months from the index date. Patients were followed from the index date to the earliest of disenrollment, death, or the study end.

Study population

- Patients were classified into three cohorts:
 - Patients with **probable MASH** were identified with an adapted metabolic dysfunction-associated steatotic liver disease (MASLD) Decision Tree[3] (hepatic steatosis index >36 or ICD-10-CM code K76.0) plus ≥1 cardiometabolic risk factor.
 - Patients with **diagnosed MASH** required ≥1 primary inpatient or ≥2 outpatient codes (K75.81) ≥30 days apart.
 - Patients **without MASH** without a MASH diagnosis and no evidence of MASLD.
- Patients were excluded if, during the baseline period, they had pre-specified liver and systemic conditions, primary or metastatic malignancy (except non-melanoma skin cancer), any open claims during the study period, or age <35 years at index date.

Study outcomes

- HCRU and costs in the follow-up period were analyzed, including all-cause inpatient visits, outpatient visits, emergency department (ED) visits, and prescriptions.
- Major adverse liver outcomes (MALO)-related, and major adverse cardiovascular events (MACE)-related HCRU and cost were also assessed.
- Subgroup analysis evaluated the effect of baseline Fibrosis-4 (FIB-4) category (low-risk, indeterminate-risk, or high-risk; **Table 1**) on HCRU and costs among patients with probable MASH.[4,5]

Table 1. Risk categories based on FIB-4 index.

	Patients with diabetes	Patients without diabetes	
		Age < 65 years	Age ≥65 years
Low-risk	<1.00	<1.30	<2.00
Indeterminate-risk	1.0-2.67	1.30-2.67	2.00-2.67
High-risk	>2.67	>2.67	>2.67

FIB-4, Fibrosis-4

Methods (Continued)

Statistical Methods

- Patients with probable MASH were compared to those with diagnosed MASH and those without MASH, separately.
- Propensity score weighting was used to adjust for baseline differences. Covariates included age, sex, race/ethnicity, region, FIB-4, type 2 diabetes (T2D), and hypertension. Comparison between probable and diagnosed MASH cohorts additionally included categorical BMI.
- Covariate balance was assessed via standardized mean differences (SMD), with absolute SMD <0.1 indicating balance.
- HCRU and costs were reported for each cohort as per patient per year (PPPY).
- Study outcomes were evaluated using weighted generalized linear or two-part regression models, as appropriate.

Results

Patient Cohorts

- The unweighted sample included patients with probable MASH (n=1,549,485), diagnosed MASH (n=15,000), and without MASH (n=513,862).

Baseline characteristics before propensity score weighting

- The patients without MASH were on average older than patients with probable MASH; probable and diagnosed MASH cohorts showed similar age profiles **(Table 2)**.
- The patients with probable MASH had a higher proportion of individuals with T2D than those without MASH; patients with diagnosed MASH had a higher proportion of individuals with T2D than those with probable MASH. Similar comorbidity profile was shown for hypertension.

Healthcare Resource Utilization

- Across all utilization categories (inpatient, outpatient, ED, prescription), rate ratios for all-cause HCRU were significantly higher for patients with probable MASH compared to those without MASH. Patients with probable MASH had a 14% increased rate of inpatient admissions, a 12% increased rate of outpatient admissions, a 23% increased rate of ED visits and a 37% increased rate of prescription fills **(Figure 1)**.
- MALO-related and MACE-related HCRU were 27% and 35% higher among patients with probable MASH than those without MASH, respectively.
- Patients with probable MASH had 17% lower all-cause HCRU than patients with diagnosed MASH, including 88% lower MALO-related, and 24% lower MACE-related HCRU **(Figure 2).**

Costs

- All-cause costs PPPY were higher in patients with probable MASH (mean: $18,562) than in patients without MASH ($14,611) **(Figure 1)**, but lower than in patients with diagnosed MASH ($26,624) **(Figure 2)**.
- The all-cause mean cost ratio (MCR) for patients with probable MASH versus those without MASH was 1.27 (95% CI: 1.26–1.28) and 0.70 (95% CI: 0.67–0.72) versus patients with diagnosed MASH.
- MALO-related costs (MCR: 1.25; 95% CI:1.21–1.29) and MACE-related costs (MCR: 1.38; 95% CI: 1.35–1.40) were also higher among patients with probable MASH versus those without MASH.
- Patients with probable MASH had lower MALO-related costs (MCR: 0.19; 95% CI: 0.17–0.21) and MACE-related costs (MCR: 0.76; 95% CI: 0.75–0.78) versus patients with diagnosed MASH.

Subgroup analysis

- Among patients with probable MASH, patients with high-risk FIB-4 incurred significantly higher all-cause, MALO-, and MACE-related HCRU and costs than patients with low-risk FIB-4 scores. This was similarly observed among patients with indeterminate-risk FIB-4 versus patients with low-risk FIB-4 scores **(Figure 3)**.

Table 2. Selected baseline characteristics before propensity score weighting.

	Probable MASH (n = 1,549,485)	Without MASH (n = 513,862)	Diagnosed MASH (n = 15,000)
Age at index			
Mean (SD)	57.96 (12.15)	60.26 (13.93)	57.27 (11.13)
Sex, n (%)			
Female	867,651 (56.0%)	318,865 (62.1%)	9,001 (60.0%)
Male	681,834 (44.0%)	194,997 (37.9%)	5,999 (40.0%)
Race/Ethnicity, n (%)			
Asian and Pacific Islander	94,636 (6.1%)	60,245 (11.7%)	1,134 (7.6%)
Black or African American	176,537 (11.4%)	38,806 (7.6%)	764 (5.1%)
Hispanic or Latino	262,693 (17.0%)	61,064 (11.9%)	3,428 (22.9%)
Other	59,752 (3.9%)	22,846 (4.4%)	573 (3.8%)
White	873,817 (56.4%)	302,562 (58.9%)	8,388 (55.9%)
Unknown	82,050 (5.3%)	28,339 (5.5%)	713 (4.8%)
Geographic Region, n (%)			
Northeast	460,189 (29.7%)	153,551 (29.9%)	5,186 (34.6%)
Midwest	173,573 (11.2%)	50,267 (9.8%)	1,413 (9.4%)
South	536,044 (34.6%)	166,122 (32.3%)	4,726 (31.5%)
West	378,863 (24.5%)	143,692 (28.0%)	3,668 (24.5%)
Unknown	816 (0.1%)	230 (0.0%)	7 (0.0%)
FIB-4 Score, n (%)			
Low risk	1,292,160 (83.4%)	343,812 (66.9%)	8,530 (56.9%)
Indeterminate risk	212,714 (13.7%)	130,019 (25.3%)	3,660 (24.4%)
High risk	44,611 (2.9%)	40,031 (7.8%)	2,810 (18.7%)
T2D, n (%)	755,660 (48.8%)	122,833 (23.9%)	10,277 (68.5%)
Hypertension, n (%)	1,281,465 (82.7%)	323,299 (62.9%)	13,046 (87.0%)
BMI, n (%)			
Normal/Underweight	83,277 (5.4%)	327,907 (63.8%)	868 (5.8%)
Overweight	478,507 (30.9%)	179,628 (35.0%)	3,141 (20.9%)
Obesity	987,701 (63.7%)	6,327 (1.2%)	10,991 (73.3%)

BMI, body mass index; **FIB-4**, fibrosis-4; **MASH**, metabolic dysfunction-associated steatohepatitis; **SD**, standard deviation; **T2D,** type-2 diabetes

Note: After weighting, baseline demographics and key clinical characteristics were balanced across the comparison groups (Without MASH effective sample size [ESS] = 247,662; Diagnosed MASH ESS = 7,537).

Figure 3. HCRU and costs comparisons between FIB-4 categories among probable MASH (subgroup analysis).

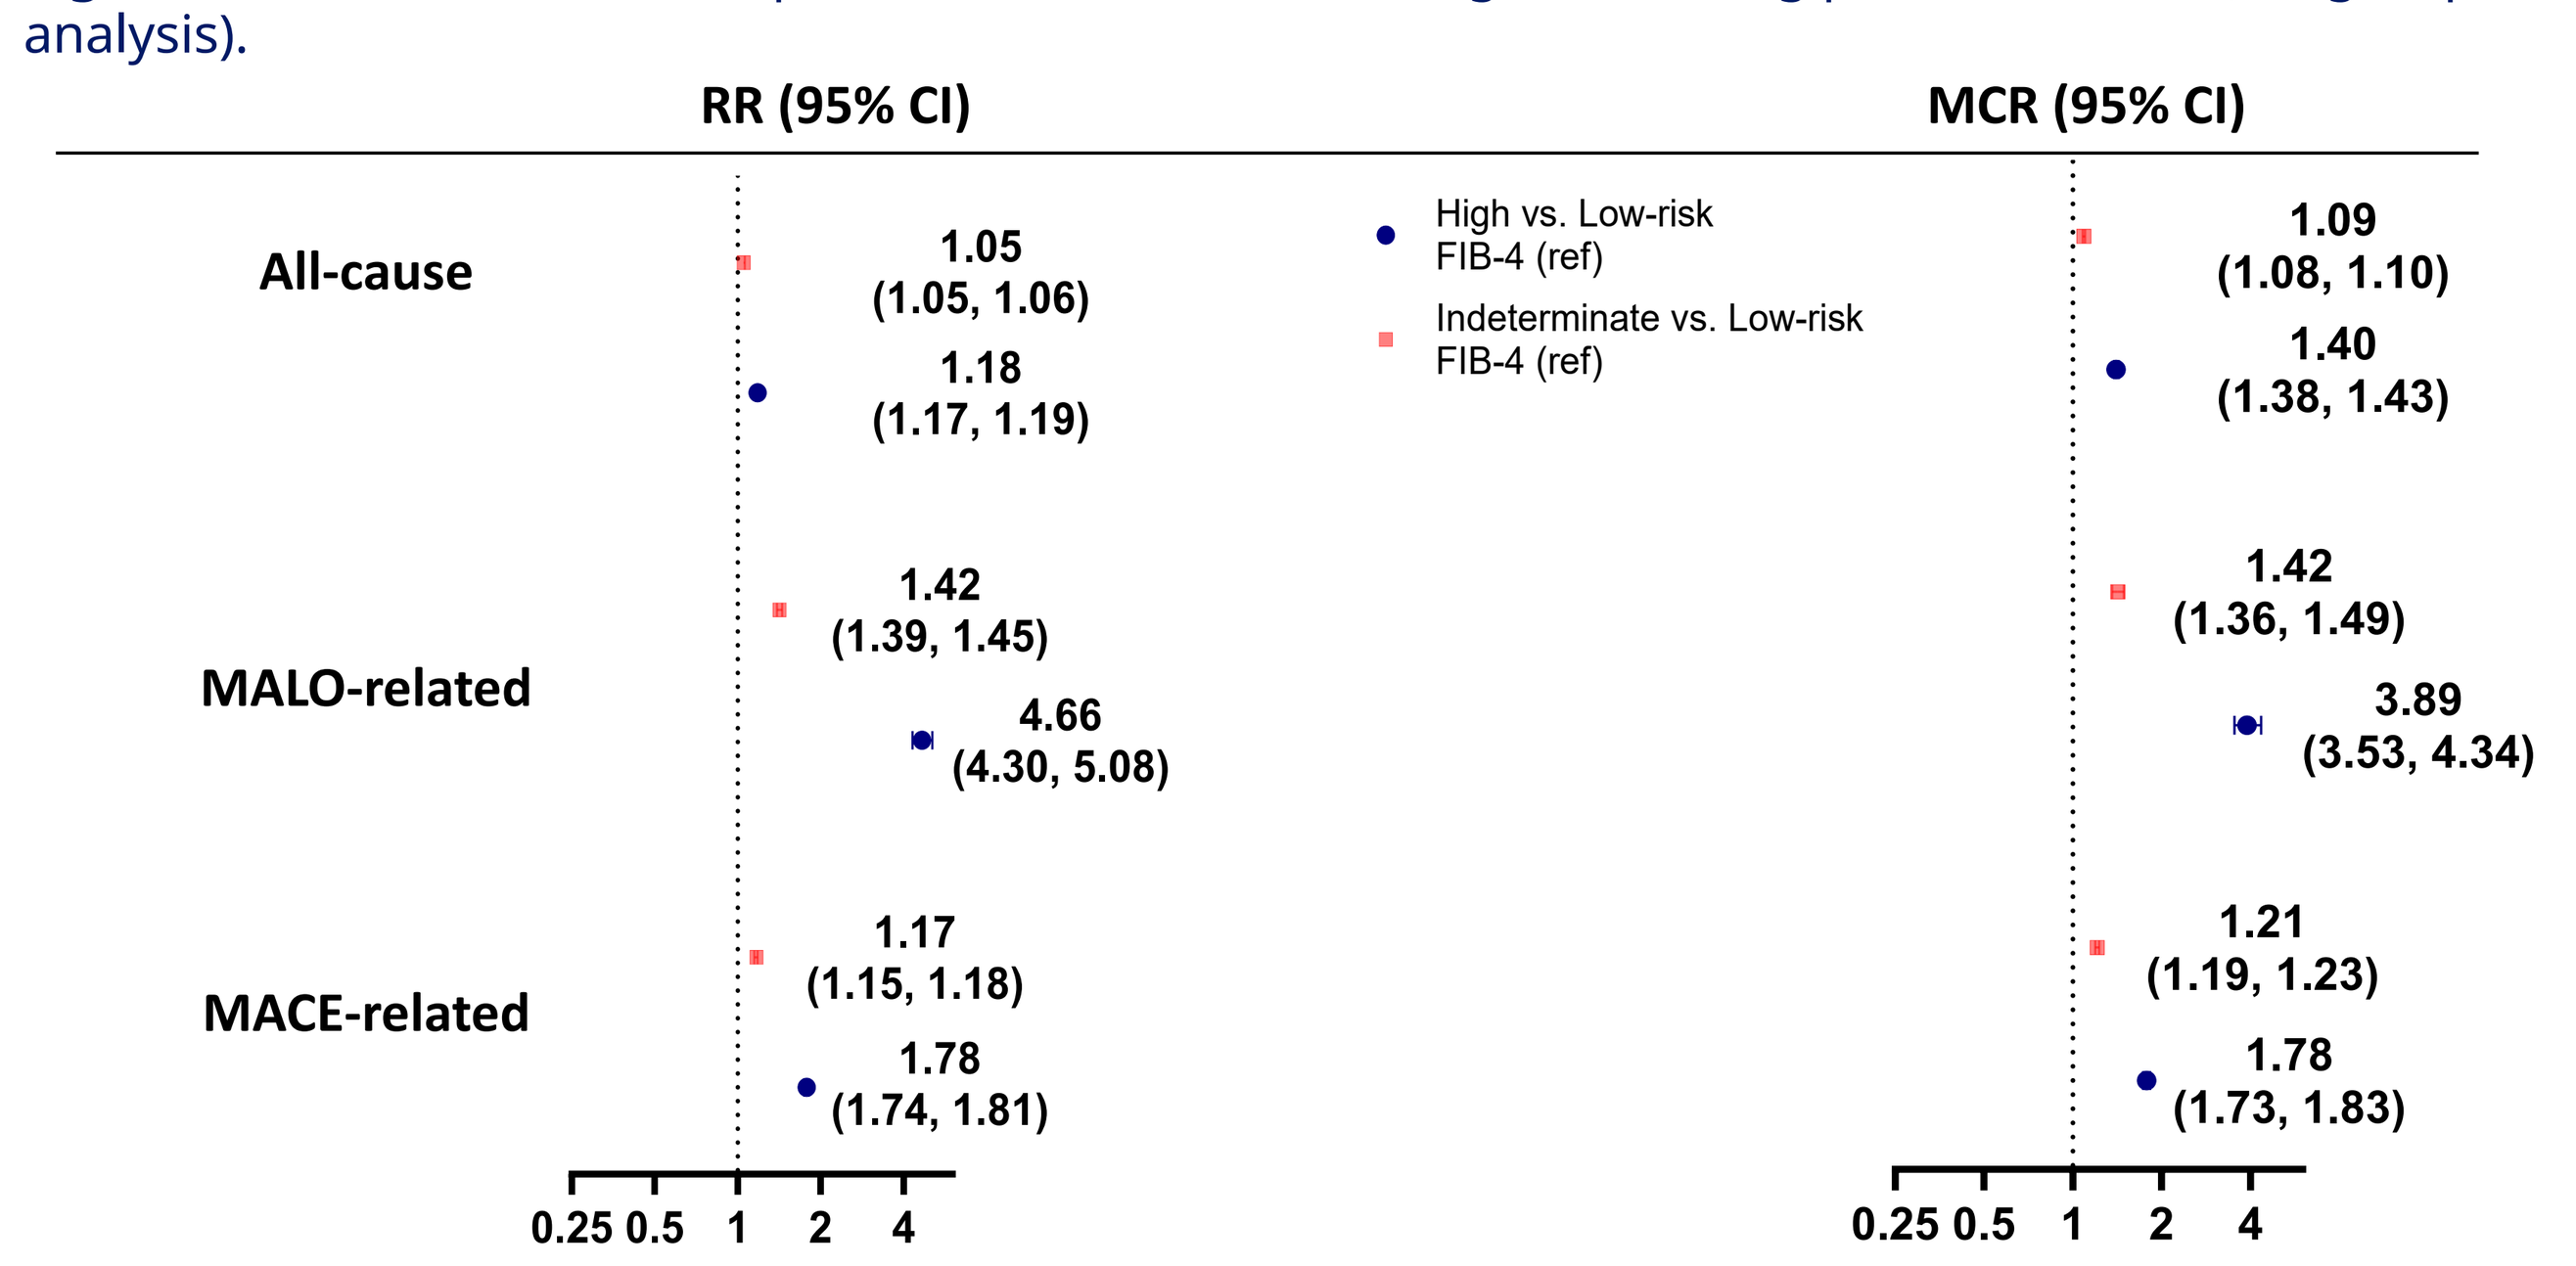

RR (95% CI)
MCR (95% CI)
High vs. Low-risk FIB-4 (ref)
Indeterminate vs. Low-risk FIB-4 (ref)
All-cause
1.05 (1.05, 1.06)
1.18 (1.17, 1.19)
1.09 (1.08, 1.10)
1.40 (1.38, 1.43)
MALO-related
1.42 (1.39, 1.45)
4.66 (4.30, 5.08)
1.42 (1.36, 1.49)
3.89 (3.53, 4.34)
MACE-related
1.17 (1.15, 1.18)
1.78 (1.74, 1.81)
1.21 (1.19, 1.23)
1.78 (1.73, 1.83)
0.25 0.5 1 2 4

CI, confidence interval; **HCRU**, healthcare resource utilization; **MACE**, major adverse cardiovascular event; **MALO,** major adverse liver outcomes; **MASH**, metabolic dysfunction-associated steatohepatitis; **RR**, rate ratio; **MCR**, mean cost ratio

Figure 1. HCRU and costs comparisons between probable MASH and without MASH (reference) cohorts after PS weighting.

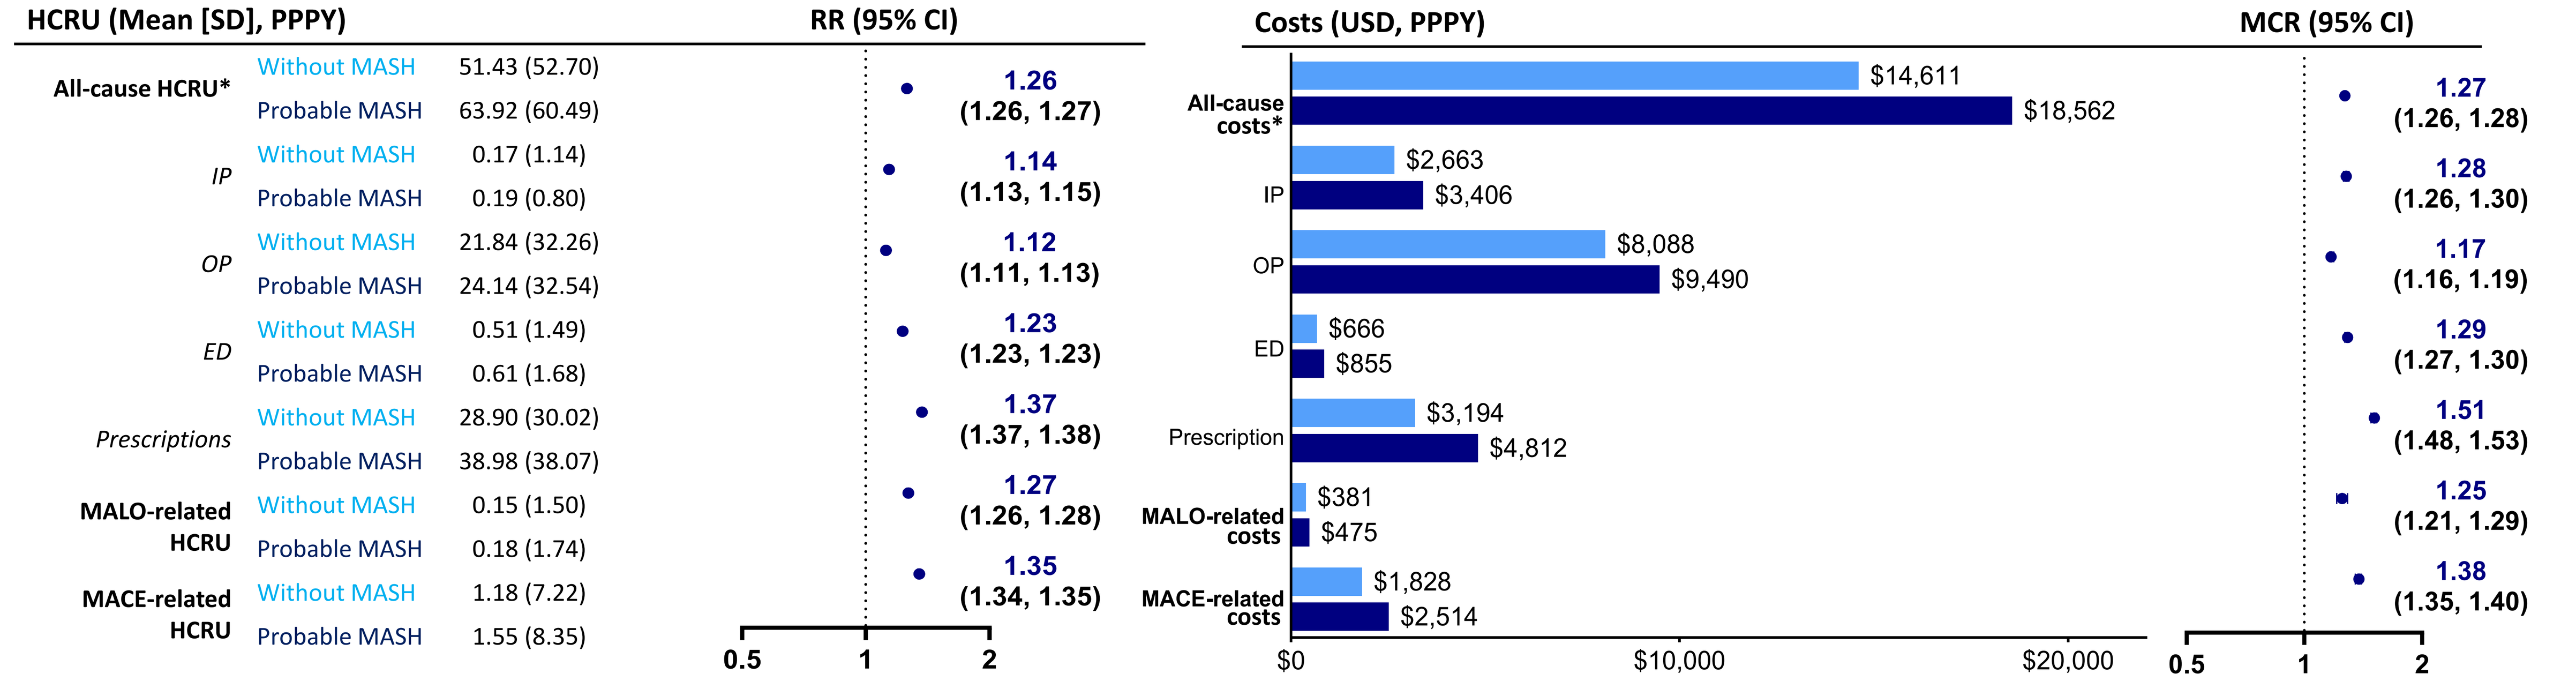

HCRU (Mean [SD], PPPY)
RR (95% CI)
Costs (USD, PPPY)
MCR (95% CI)
All-cause HCRU*
Without MASH 51.43 (52.70)
Probable MASH 63.92 (60.49)
1.26 (1.26, 1.27)
IP
Without MASH 0.17 (1.14)
Probable MASH 0.19 (0.80)
1.14 (1.13, 1.15)
OP
Without MASH 21.84 (32.26)
Probable MASH 24.14 (32.54)
1.12 (1.11, 1.13)
ED
Without MASH 0.51 (1.49)
Probable MASH 0.61 (1.68)
1.23 (1.23, 1.23)
Prescriptions
Without MASH 28.90 (30.02)
Probable MASH 38.98 (38.07)
1.37 (1.37, 1.38)
MALO-related HCRU
Without MASH 0.15 (1.50)
Probable MASH 0.18 (1.74)
1.27 (1.26, 1.28)
MACE-related HCRU
Without MASH 1.18 (7.22)
Probable MASH 1.55 (8.35)
1.35 (1.34, 1.35)
0.5 1 2
All-cause costs* $14,611 $18,562
IP $2,663 $3,406
OP $8,088 $9,490
ED $666 $855
Prescription $3,194 $4,812
MALO-related costs $381 $475
MACE-related costs $1,828 $2,514
$0 $10,000 $20,000
1.27 (1.26, 1.28)
1.28 (1.26, 1.30)
1.17 (1.16, 1.19)
1.29 (1.27, 1.30)
1.51 (1.48, 1.53)
1.25 (1.21, 1.29)
1.38 (1.35, 1.40)
0.5 1 2

CI, confidence interval; **ED**, emergency department; **HCRU**, healthcare resource utilization; **IP**, inpatient; **MACE**, major adverse cardiovascular event; **MALO**, major adverse liver outcomes; **MASH**, metabolic dysfunction-associated steatohepatitis; **MCR**, mean cost ratio; **OP**, outpatient; **PPPY**, per-patient per-year; **RR**, rate ratio; **SD**, standard deviation; **USD**, US dollars.

Note: *All-cause HCRU/costs includes IP, OP, ED visits, and prescription fills.

Figure 2. HCRU and costs comparisons between probable MASH and diagnosed MASH (reference) cohorts after PS weighting.

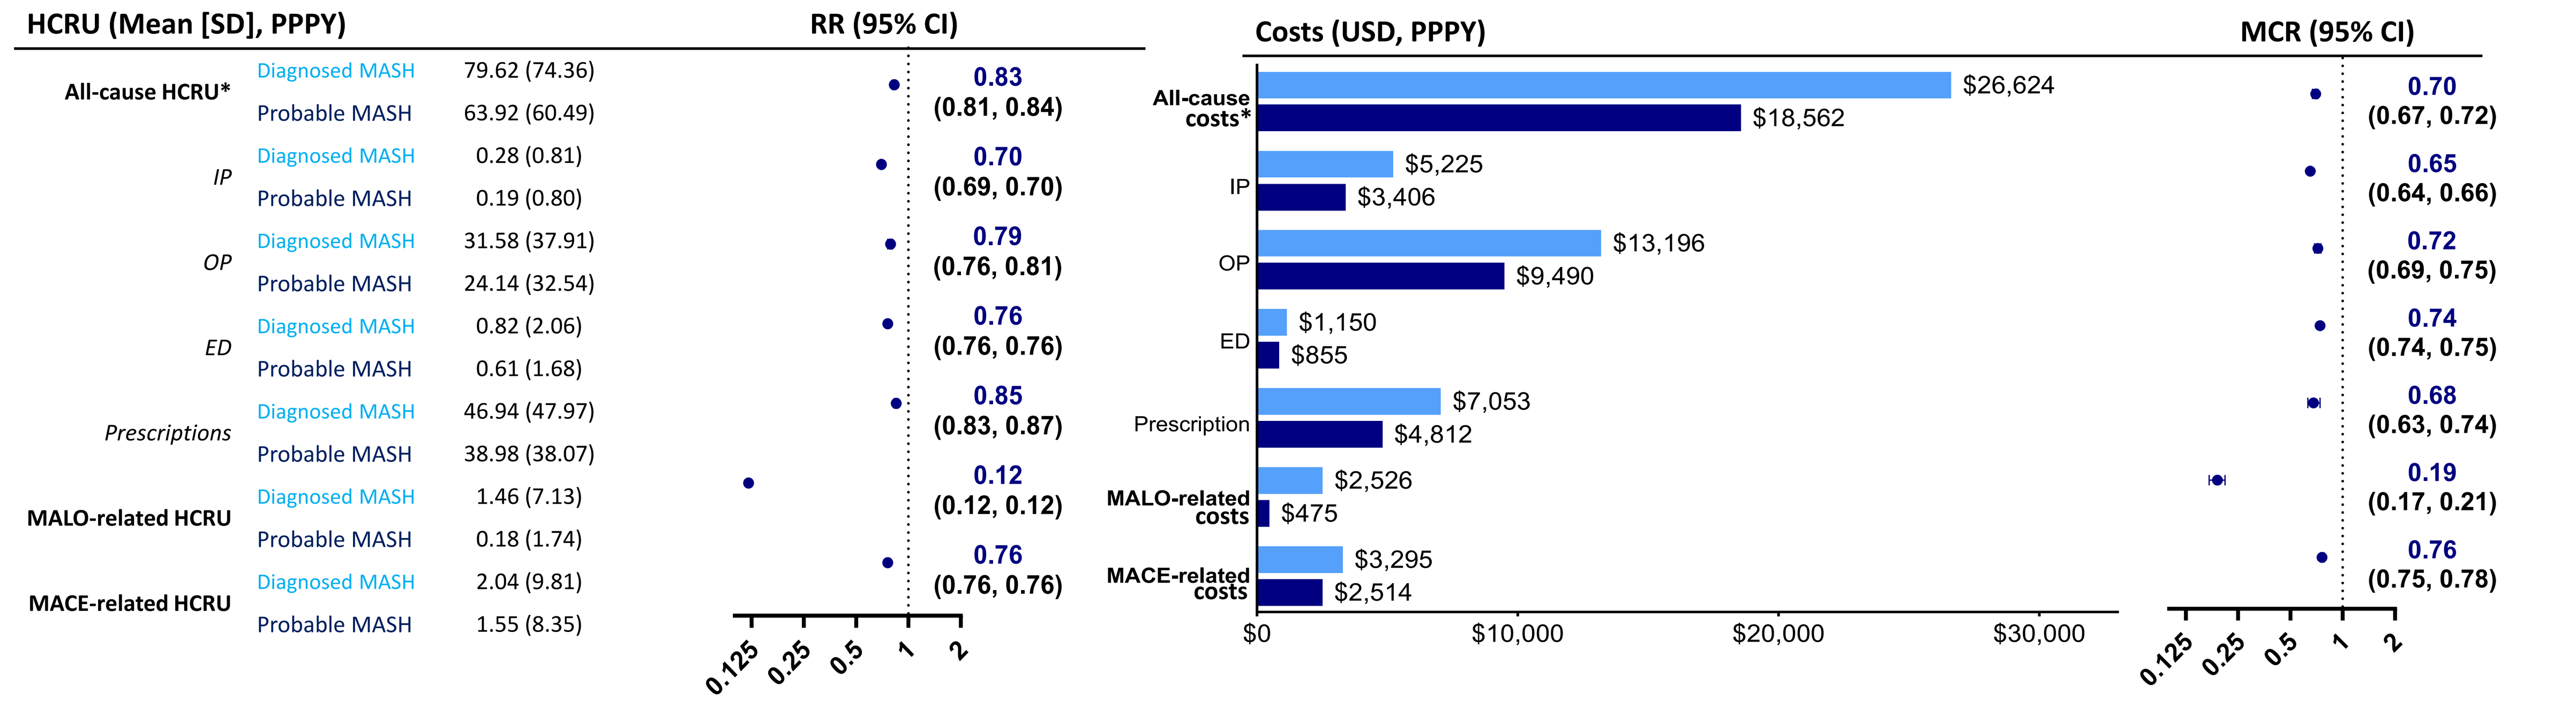

HCRU (Mean [SD], PPPY)
RR (95% CI)
Costs (USD, PPPY)
MCR (95% CI)
All-cause HCRU*
Diagnosed MASH 79.62 (74.36)
Probable MASH 63.92 (60.49)
0.83 (0.81, 0.84)
IP
Diagnosed MASH 0.28 (0.81)
Probable MASH 0.19 (0.80)
0.70 (0.69, 0.70)
OP
Diagnosed MASH 31.58 (37.91)
Probable MASH 24.14 (32.54)
0.79 (0.76, 0.81)
ED
Diagnosed MASH 0.82 (2.06)
Probable MASH 0.61 (1.68)
0.76 (0.76, 0.76)
Prescriptions
Diagnosed MASH 46.94 (47.97)
Probable MASH 38.98 (38.07)
0.85 (0.83, 0.87)
MALO-related HCRU
Diagnosed MASH 1.46 (7.13)
Probable MASH 0.18 (1.74)
0.12 (0.12, 0.12)
MACE-related HCRU
Diagnosed MASH 2.04 (9.81)
Probable MASH 1.55 (8.35)
0.76 (0.76, 0.76)
0.125 0.25 0.5 1 2
All-cause costs* $26,624 $18,562
IP $5,225 $3,406
OP $13,196 $9,490
ED $1,150 $855
Prescription $7,053 $4,812
MALO-related costs $2,526 $475
MACE-related costs $3,295 $2,514
$0 $10,000 $20,000 $30,000
0.70 (0.67, 0.72)
0.65 (0.64, 0.66)
0.72 (0.69, 0.75)
0.74 (0.74, 0.75)
0.68 (0.63, 0.74)
0.19 (0.17, 0.21)
0.76 (0.75, 0.78)
0.125 0.25 0.5 1 2

CI, confidence interval; **ED**, emergency department; **HCRU**, healthcare resource utilization; **IP**, inpatient; **MACE**, major adverse cardiovascular event; **MALO**, major adverse liver outcomes; **MASH**, metabolic dysfunction-associated steatohepatitis; **MCR**, mean cost ratio; **OP**, outpatient; **PPPY**, per-patient per-year; **RR**, rate ratio; **SD**, standard deviation; **USD**, US dollars.

Note: *All-cause HCRU/costs includes IP, OP, ED visits, and prescription fills.

Limitations

- Administrative claims data are subject to coding errors, potentially misclassifying MASH, covariates, HCRU or costs, and leading to possible under/overreporting of events.
- Identification of patients with probable MASH may lack precision and could potentially represent a cohort of patients primarily with MASLD; however, this should be addressed by the subgroup analysis of patients with probable MASH based on their FIB-4 risk category.

Conclusion

- A substantial number of patients with probable MASH were identified, and our study findings highlight this population's significant economic burden (All-cause costs PPPY: $18,562).
- Patients with probable MASH incurred significantly higher HCRU and costs than patients without MASH, but lower HCRU and costs than patients with diagnosed MASH.
- Among patients with probable MASH, patients with high/indeterminate risk FIB-4 category incurred significantly higher HCRU and costs than patients with low-risk FIB-4 category.
- These findings demonstrate the importance of early diagnosis and intervention for patients with MASH.
- There is a general need for early identification among patients with MASH, and this can potentially reduce the economic burden associated with this underdiagnosed patient population.

The authors thank Sonia Kim and Yingjie Ding of Genesis Research Group, Hoboken, NJ, US, who provided medical writing and editorial support.

Presented at the AMCP Nexus 2025 conference, October 27–30, National Harbor, MD, USA.

Disclosures:
CZ, SOG, MJ, AA, and RL are employees of Novo Nordisk Inc. and shareholders of Novo Nordisk A/S. NB, AS and BZ are employees of Genesis Research Group which was contracted for the conduct of this study. MER is a consultant/advisor to Novo Nordisk Inc.

References:
(1) Charlton M, et al. *J Manag Care Spec Pharm*. 2024;30:1414–1430; (**2**) Kim Y, et al. *Liver Int*. 2025;45:e70225; (**3**) Rinella ME, et al. *J Hepatol*. 2023;79:1542–1556; **(4)** McPherson S et al., Hepatology. 2017 Feb;65(2):544-553. DOI: 10.1002/hep.28843; **(5)** Rinella ME, et al. AASLD practice guidance on NAFLD. Hepatology. 2023;77:1797–1835.