Real-World Healthcare Resource Utilization and Costs Among 100% Fee-For-Service Medicare Beneficiaries With Metabolic Dysfunction-Associated Steatohepatitis

Husam Albarmawi[1]; Anran Tan[2]; Abdalla Aly[1]; Anthony Hoovler[1]; Caichen Zhong[1]; Jamieson Vaccaro[2]; Joanna Harton[2]; Parvez Mantry[3].
[1]Novo Nordisk Inc., Plainsboro, NJ, USA; [2]Genesis Research Group, Hoboken, NJ, USA; [3]Methodist Dallas Medical Center, Dallas, TX, USA.

Aims

Estimate the incremental healthcare resource utilization (HCRU) and cost burden of metabolic dysfunction-associated steatohepatitis (MASH) and cirrhotic MASH among Medicare beneficiaries:

1. compare those diagnosed with MASH vs those never diagnosed with MASH;
2. compare those diagnosed with cirrhotic MASH vs those with non-cirrhotic MASH.

Introduction

- Metabolic dysfunction-associated steatotic liver disease (MASLD) is characterized by the abnormal accumulation of lipids in the liver and is the leading cause of chronic liver disease in the US.
- It is estimated that within 8-13 years 12-40% of patients with MASLD progress to a more severe form of the disease,[1] MASH, formerly known as nonalcoholic steatohepatitis (NASH).
- Prior studies have evaluated HCRU and costs incurred by patients with MASH as well as the incremental burden associated with cirrhotic MASH,[2] MASH progression,[3,4] and fibrosis stage.[5,6]
- Evidence regarding the healthcare burden of MASH remains limited for: (a) Medicare beneficiaries and (b) incremental estimates comparing MASH with non-MASH populations.

Methods

Study design and data source

- This was a non-interventional, retrospective cohort study of Medicare beneficiaries diagnosed with MASH as well as those never diagnosed with MASH to assess the impact of MASH diagnosis and severity on HCRU and costs among beneficiaries aged ≥66 years old.
- The study utilized 100% Centers for Medicare & Medicaid Services (CMS) fee-for-service (FFS) claims ranging from January 1, 2016, through December 31, 2022 (study period).
- Datasets used for the analysis included Medicare Parts A, B, and D to evaluate patient characteristics, HCRU, and healthcare costs related to medical services and pharmacy use.
- The index date for the non-MASH cohort was the first documented claim in the identification period with ≥12 months of continuous enrollment (eligibility date). For other cohorts, the index date was the later of eligibility and: a) the earliest of first MASH or cirrhosis diagnosis for the MASH cohort; b) the first cirrhosis diagnosis for the cirrhotic-MASH cohort; and c) the first MASH diagnosis for the non-cirrhotic-MASH cohort.
- The baseline period was set as the 12 months preceding the cohort-specific index date.
- The follow-up period was defined as the time from the cohort-specific index date until the first occurrence of disenrollment, health maintenance organization (HMO) enrollment, Medicaid eligibility, death, or the end of the study.

Study population

- The study population included 100% CMS FFS beneficiaries aged ≥66 years old from January 1, 2017, through December 31, 2022 (identification period).
- After inclusion of beneficiaries who met the general study inclusion criteria, the study population was divided into 2 main cohorts: (a) all beneficiaries diagnosed with MASH during the study period and (b) a random 5:1 sample (relative to the MASH cohort) of beneficiaries never diagnosed with MASH during the study period. The MASH cohort was further divided into 2 sub-cohorts: (c) patients diagnosed with cirrhosis during the study period; (d) patients never diagnosed with cirrhosis. **(Figure 1)**
- The final cohorts were obtained after exclusion of beneficiaries who met cohort-specific and general study exclusion criteria. **(Figure 1)**

Study outcomes

- All-cause HCRU was reported as the number of visits/fills per-patient per-year (PPPY) for the following categories: inpatient (IP), outpatient (OP), emergency room (ER), skilled nursing facility and hospice (SNF/H), and prescription fills.
- All-cause costs PPPY were reported by category, with totals for medical (IP + OP + ER + SNF/H) and overall costs, in 2022 US dollars (Consumer Price Index).

Statistical methods

- Stabilized inverse probability weighting (IPW) was used to adjust for differences in baseline covariates, including differences in cardiometabolic conditions, between comparison cohorts.
- Baseline characteristics prior to IPW were summarized descriptively, and covariate balance between comparison cohorts was quantified by standardized mean differences (SMDs).
- Post-IPW HCRU and costs were reported with 95% Poisson exact confidence intervals (CIs).
- Ratios and differences in HCRU incidence and costs rates between comparison cohorts were estimated with 95% CIs using univariate weighted generalized linear models.

Results

Patient attrition

- 128,622 beneficiaries were included in the MASH cohort, 642,774 in the non-MASH cohort, 52,883 in the cirrhotic MASH cohort and 74,910 in the non-cirrhotic MASH cohort. **(Figure 1)**

Baseline characteristics before IPW

- Beneficiaries with MASH were modestly younger than those without MASH (mean age: 73.4 vs 74.7 years) and included a slightly higher percentage of females (60.4% vs 56.8%). **(Table)**
- Beneficiaries with MASH were marked by a substantially greater Charlson Comorbidity Index (3.7 vs 1.7) and were more often diagnosed with cardiometabolic conditions. **(Table)**
- Beneficiaries with cirrhotic MASH were slightly older than those with non-cirrhotic MASH (mean age: 74.1 vs 73.1 years) and included a marginally lower percentage of females (59.6% vs 61.1%). **(Table)**
- Beneficiaries with cirrhotic MASH were marked by a considerably greater Charlson Comorbidity Index (4.9 vs 2.8) and were more often diagnosed with type 2 diabetes, atherosclerotic cardiovascular disease, and heart failure. **(Table)**

Baseline characteristics after IPW

- The effective sample sizes (ESS) were 52,845 for the MASH and 543,283 for the non-MASH cohorts, and their adjusted baseline covariates were well balanced (SMD <0.1). *(Data not shown)*
- The ESS were 37,607 for the cirrhotic MASH and 49,268 for the non-cirrhotic MASH cohorts, and their adjusted baseline covariates were well balanced (SMD <0.1). *(Data not shown)*

Figure 1. Patient attrition.

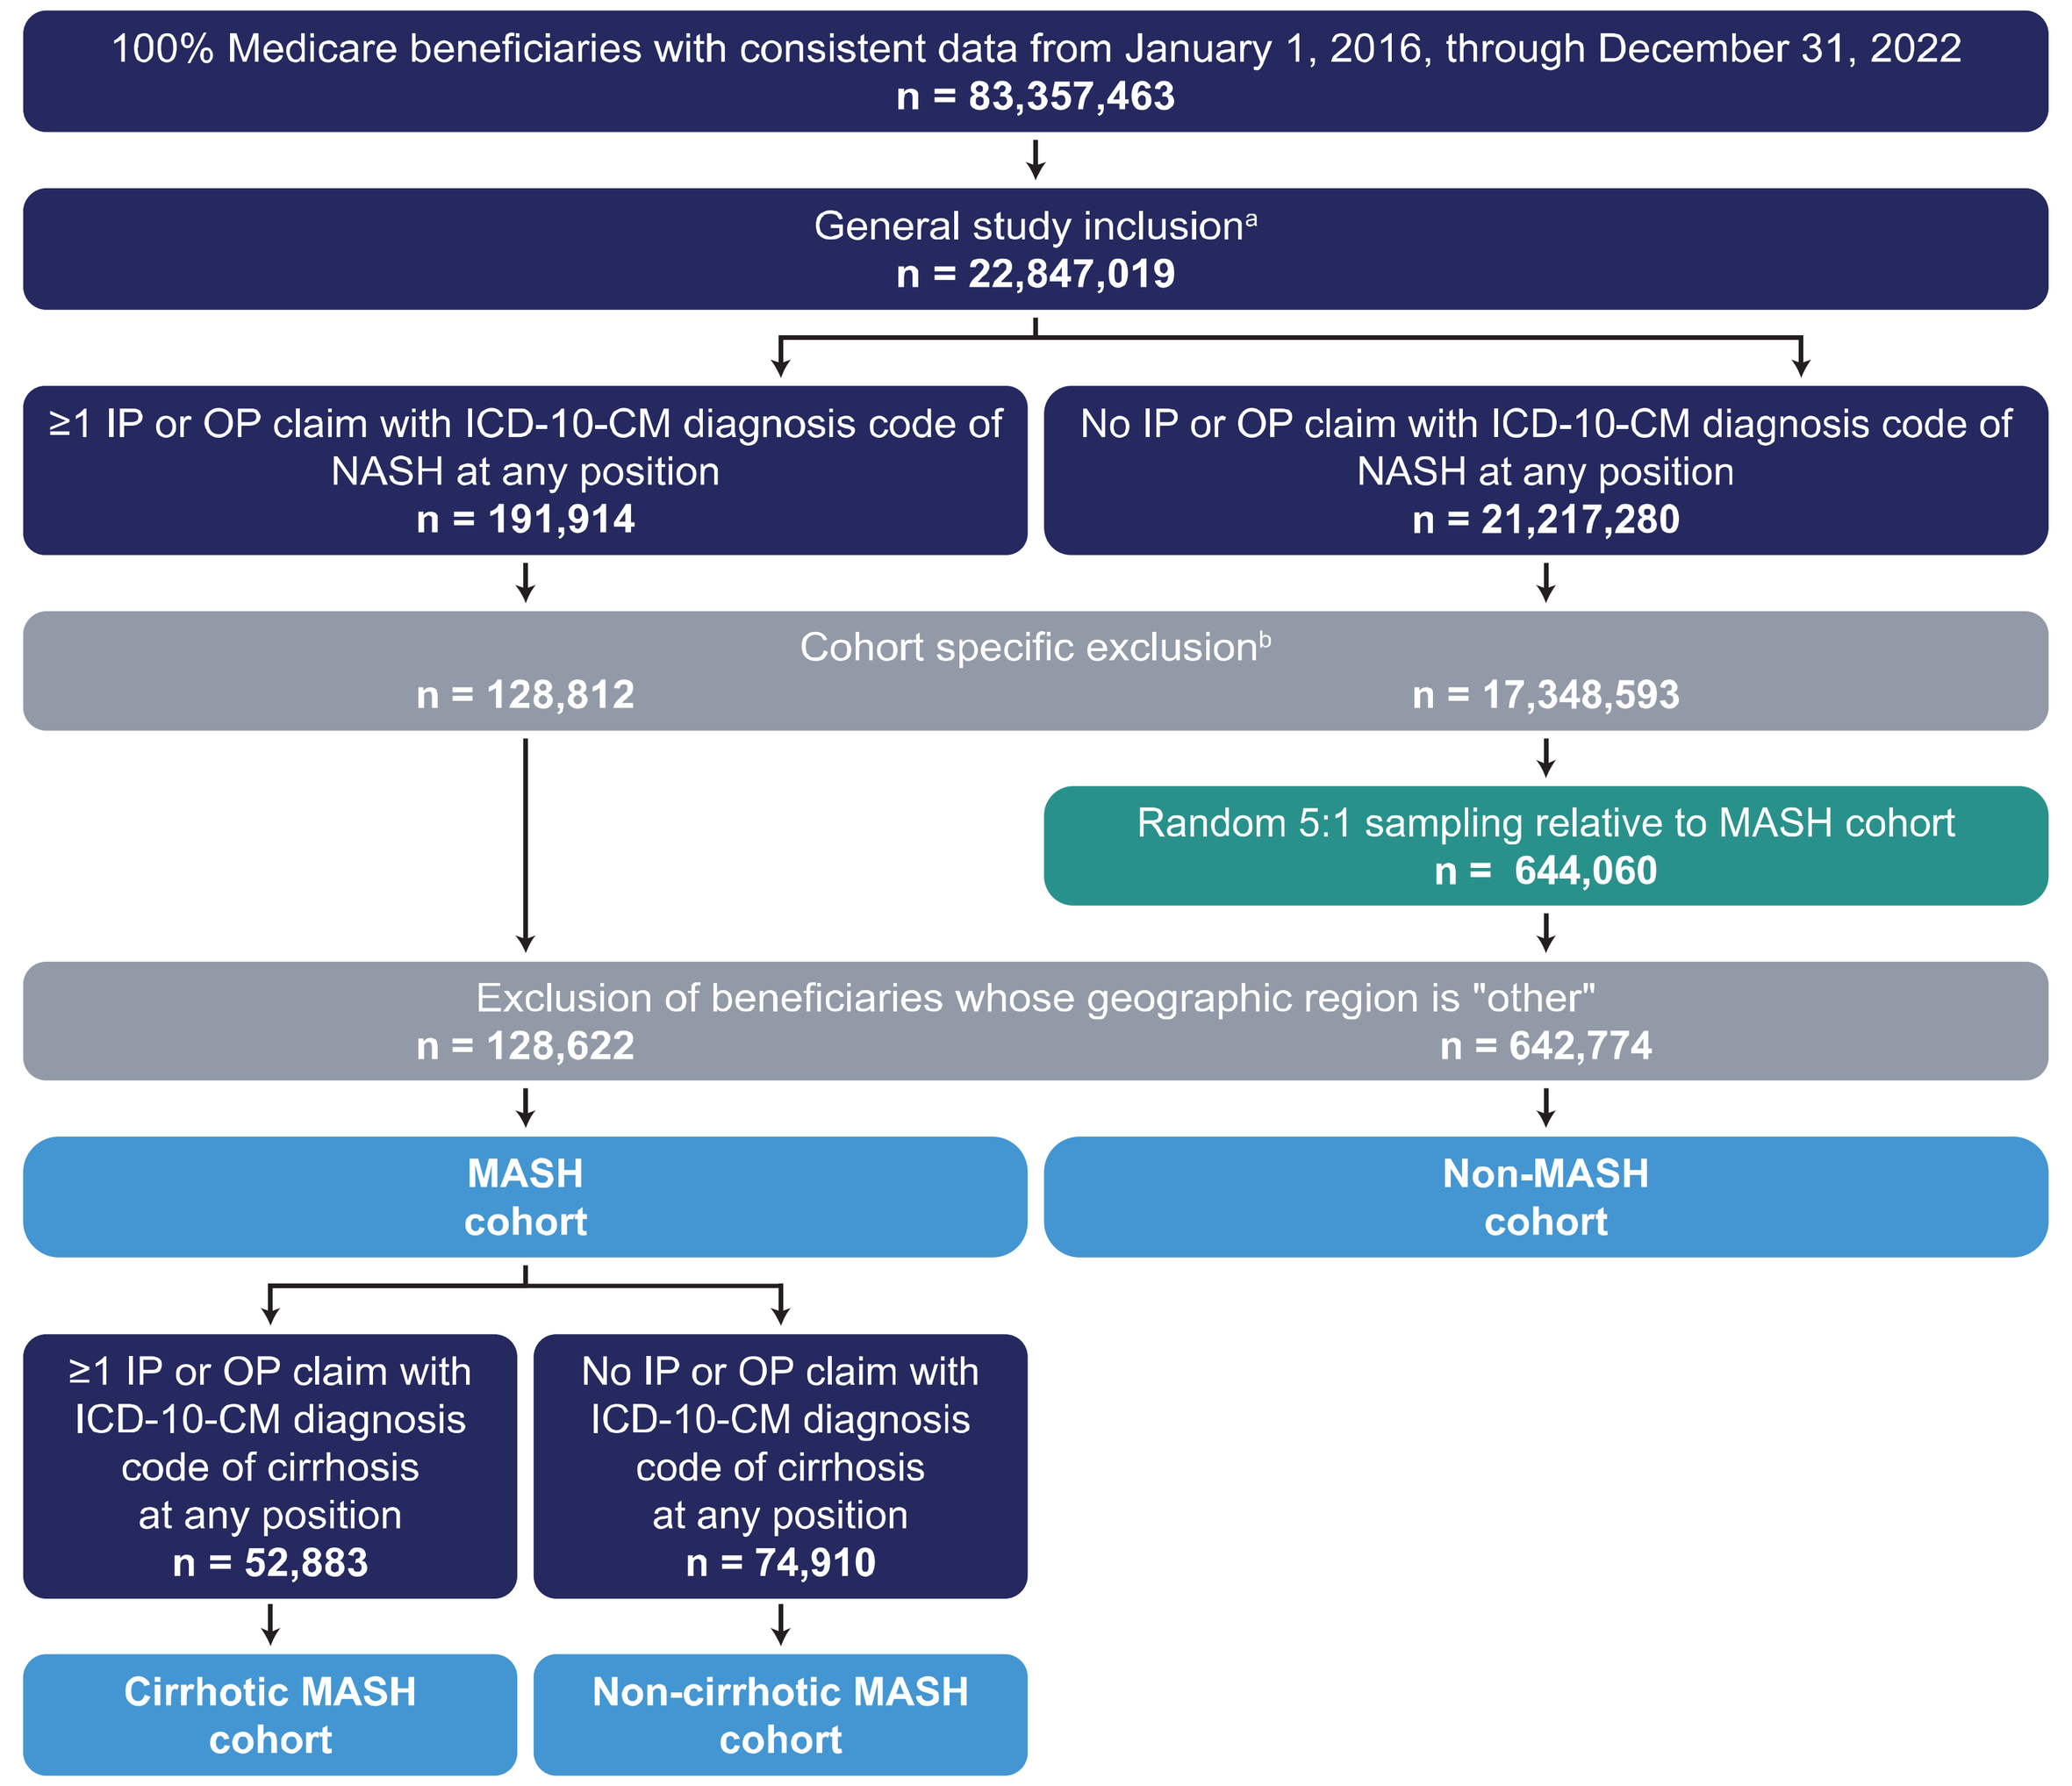
100% Medicare beneficiaries with consistent data from January 1, 2016, through December 31, 2022
n = 83,357,463
General study inclusion[a]
n = 22,847,019
≥1 IP or OP claim with ICD-10-CM diagnosis code of NASH at any position
n = 191,914
No IP or OP claim with ICD-10-CM diagnosis code of NASH at any position
n = 21,217,280
Cohort specific exclusion[b]
n = 128,812
n = 17,348,593
Random 5:1 sampling relative to MASH cohort
n = 644,060
Exclusion of beneficiaries whose geographic region is "other"
n = 128,622
n = 642,774
MASH cohort
Non-MASH cohort
≥1 IP or OP claim with ICD-10-CM diagnosis code of cirrhosis at any position
n = 52,883
No IP or OP claim with ICD-10-CM diagnosis code of cirrhosis at any position
n = 74,910
Cirrhotic MASH cohort
Non-cirrhotic MASH cohort

CMS, Centers for Medicare & Medicaid Services; FFS, fee-for-service; HMO, health maintenance organization; ICD-10-CM, International Classification of Diseases, Tenth Revision, Clinical Modification; IP, inpatient; MASH, metabolic dysfunction-associated steatohepatitis; NASH, nonalcoholic steatohepatitis; OP, outpatient.

[a] General inclusion criteria: (a) ≥12 months of continuous enrollment in traditional CMS FFS Parts A, B and D and no enrollment in an HMO; (b) were ≥65 years old at the start of their continuous enrollment period (and thus ≥66 years old at their index date).

[b] Cohort-specific exclusion criteria: (a) they had <12 months of continuous enrollment in traditional CMS FFS Parts A, B and D or were enrolled in an HMO in the year before the index date; (b) they were eligible for dual Medicare/Medicaid during the baseline period; (c) they had a record of competing conditions associated with liver injury (for MASH cohorts and sub-cohorts only); or (d) death occurred on or before the index date.

MASH vs Non-MASH HCRU and costs outcomes

- MASH diagnosis was associated with higher HCRU for all categories: all-cause PPPY HCRU was greater by 48% for IP visits (0.4 vs 0.3), 32% for ER visits (0.8 vs 0.2), 20% for both OP visits (37.5 vs 31.3) and SNF/H stays (0.22 vs 0.18), and 7% for prescription drug fills (31.8 vs 29.7). **(Figure 2)**
- Total costs PPPY were 21% higher for MASH vs non-MASH ($24,890 vs $20,542), including higher IP ($6,855 vs $4,604), OP ($11,107 vs $9,565), prescription drug ($4,634 vs $4,039), SNF/H care ($1,874 vs $1,653), and ER costs ($635 vs $498). **(Figure 2)**

Cirrhotic MASH vs Non-Cirrhotic MASH HCRU and costs outcomes

- Among patients with MASH, cirrhosis diagnosis was associated with higher HCRU for all categories: all-cause PPPY HCRU was greater by 136% for IP visits (0.9 vs 0.4), 104% for ER visits (1.6 vs 0.8), 94% for SNF/H stays (0.4 vs 0.2), 36% for OP visits (57.8 vs 42.6), and 10% for prescription drug fills (41.6 vs 37.9). **(Figure 3)**
- Total costs PPPY were 53% higher for cirrhotic MASH vs non-cirrhotic MASH ($45,050 vs $29,410), including higher IP ($14,951 vs $6,571), OP ($18,077 vs $13,580), SNF/H care ($3,337 vs $1,789), prescription fills ($7,297 vs $6,453), and ER costs ($1,166 vs $619). **(Figure 3)**

Table. Selected baseline characteristics before IPW.

	MASH (n = 128,622)	Non-MASH (n = 642,774)	SMD	Cirrhotic MASH (n = 52,883)	Non-cirrhotic MASH (n = 74,910)	SMD
Age at index						
Mean, years (SD)	73.4 (5.5)	74.7 (7.4)	0.19	74.1 (5.7)	73.1 (5.4)	0.17
Sex, n (%)			0.07			0.03
Male	50,914 (39.6%)	277,876 (43.2%)		21,377 (40.4%)	29,175 (38.9%)	
Female	77,708 (60.4%)	364,898 (56.8%)		31,506 (59.6%)	45,735 (61.1%)	
Race/Ethnicity, n (%)			0.12			0.15
White	117,777 (91.6%)	578,124 (89.9%)		49,492 (93.6%)	67,521 (90.1%)	
Black	3,332 (2.6%)	30,816 (4.8%)		887 (1.7%)	2,422 (3.2%)	
Asian	1,443 (1.1%)	6,812 (1.1%)		363 (0.7%)	1,075 (1.4%)	
Hispanic	897 (0.7%)	3,112 (0.5%)		342 (0.6%)	543 (0.7%)	
North American Native	540 (0.4%)	1,407 (0.2%)		281 (0.5%)	257 (0.3%)	
Other/Unknown	4,633 (3.6%)	22,503 (3.5%)		1,518 (2.9%)	3,092 (4.1%)	
Charlson Comorbidity Index						
Mean (SD)	3.7 (2.7)	1.7 (2.0)	0.85	4.9 (2.8)	2.8 (2.3)	0.81
Selected Comorbidities, n (%)						
Type 2 diabetes	74,974 (58.3%)	171,145 (26.6%)	0.68	36,830 (69.6%)	37,331 (49.8%)	0.41
IFG/IGT	31,491 (24.5%)	107,770 (16.8%)	0.19	9,314 (17.6%)	21,811 (29.1%)	0.27
Hyperlipidemia	104,618 (81.3%)	417,545 (65.0%)	0.38	40,438 (76.5%)	63,016 (84.1%)	0.19
Hypertension	112,014 (87.1%)	455,675 (70.9%)	0.41	46,959 (88.8%)	63,759 (85.1%)	0.11
Overweight	17,498 (13.6%)	54,230 (8.4%)	0.17	6,521 (12.3%)	10,803 (14.4%)	0.06
Obesity	58,267 (45.3%)	106,251 (16.5%)	0.66	25,220 (47.7%)	32,412 (43.3%)	0.09
Chronic kidney disease	29,518 (22.9%)	70,528 (11.0%)	0.32	15,399 (29.1%)	13,818 (18.4%)	0.25
Obstructive sleep apnea	33,035 (25.7%)	63,579 (9.9%)	0.42	13,870 (26.2%)	18,793 (25.1%)	0.03
Polycystic ovary syndrome	70 (0.1%)	116 (0.0%)	0.02	31 (0.1%)	38 (0.1%)	0.00
Heart failure	23,720 (18.4%)	59,527 (9.3%)	0.27	14,010 (26.5%)	9,432 (12.6%)	0.36
ASCVD	56,495 (43.9%)	205,301 (31.9%)	0.25	26,000 (49.2%)	29,840 (39.8%)	0.19

ASCVD, atherosclerotic cardiovascular disease; IFG, impaired fasting glucose; IGT, impaired glucose tolerance; IPW, inverse probability weighting; MASH, metabolic dysfunction-associated steatohepatitis; SD, standard deviation; SMD, standardized mean differences.

Percentage totals may not add to 100% due to rounding.

The covariates included in the MASH/no MASH propensity score model were: categorical age, race, sex, region, index year pre-2020, Charlson Comorbidity Index, type 2 diabetes, hyperlipidemia, hypertension, ASCVD, heart failure, overweight, obese, frailty score, sleep apnea, medication at baseline including anti-diabetics, anti-hypertensives, lipid lowering therapies.

The covariates included in the cirrhotic/non-cirrhotic propensity score model were: categorical age, race, sex, region, index year pre-2020, CCI, type 2 diabetes, hyperlipidemia, hypertension, ASCVD, heart failure, overweight, obese, frailty score, biliary cirrhosis, hepatitis B, hepatitis C, or other viral hepatitis, medication at baseline including anti-diabetics, anti-hypertensives, lipid lowering therapies.

Figure 2. HCRU and Costs comparisons between MASH and non-MASH cohorts after IPW.

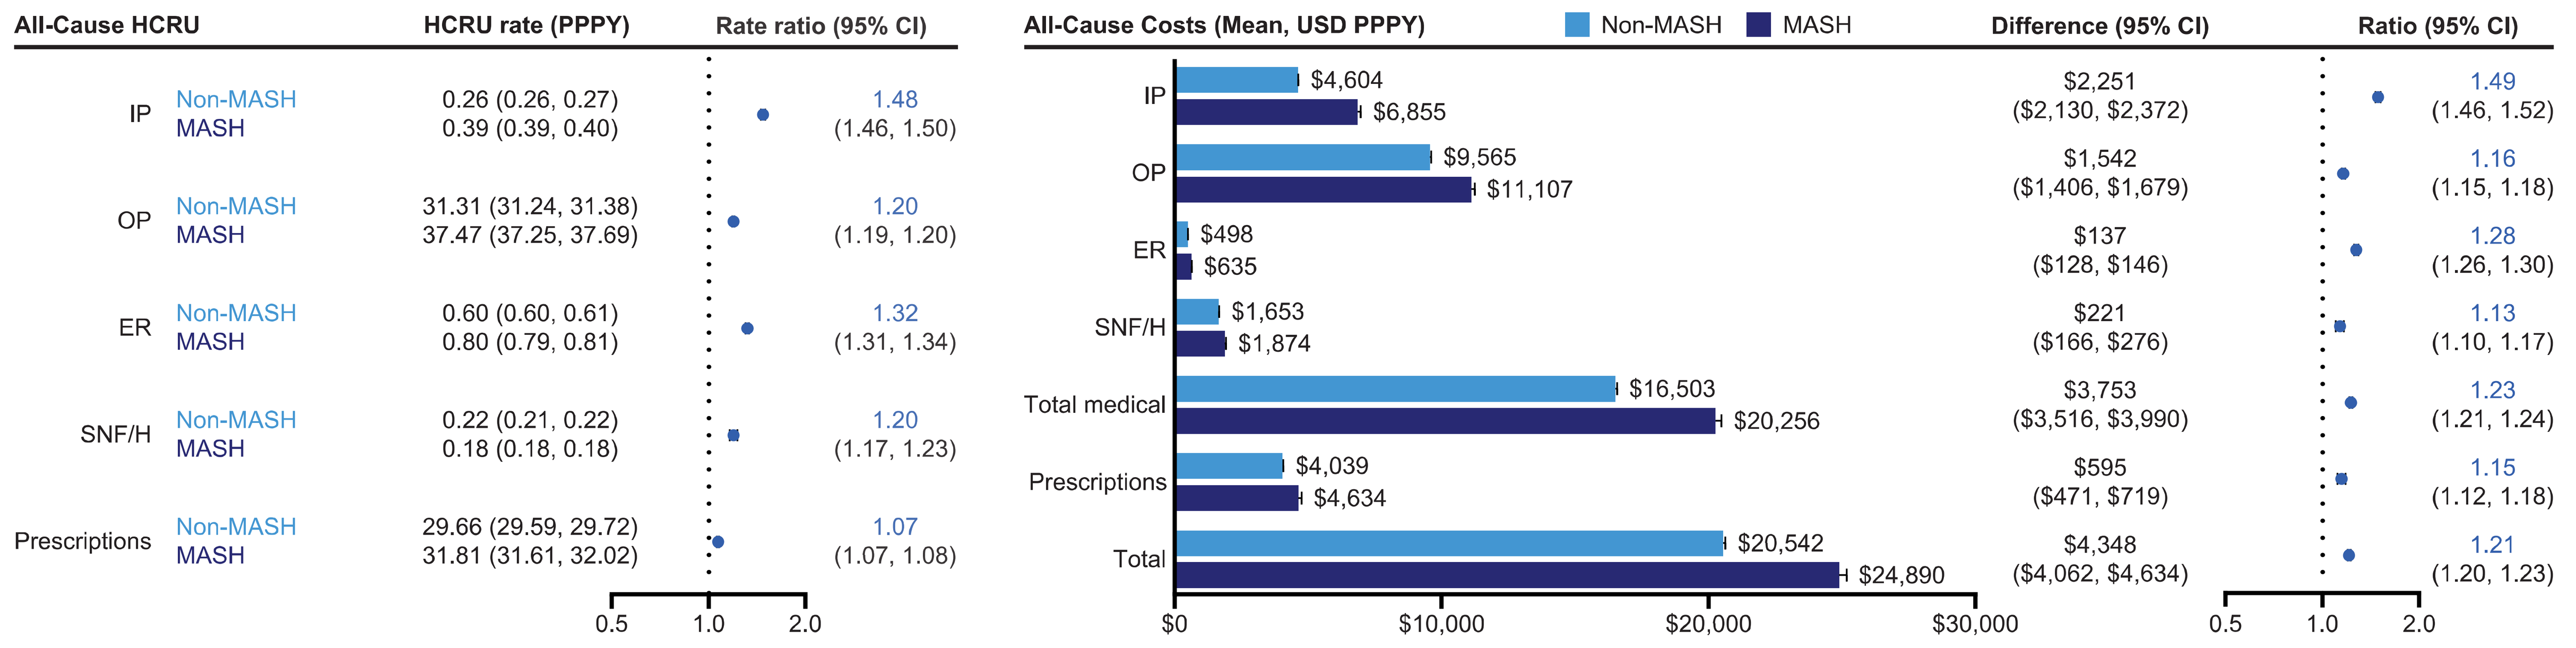
All-Cause HCRU
HCRU rate (PPPY)
Rate ratio (95% CI)
IP Non-MASH 0.26 (0.26, 0.27)
MASH 0.39 (0.39, 0.40)
1.48 (1.46, 1.50)
OP Non-MASH 31.31 (31.24, 31.38)
MASH 37.47 (37.25, 37.69)
1.20 (1.19, 1.20)
ER Non-MASH 0.60 (0.60, 0.61)
MASH 0.80 (0.79, 0.81)
1.32 (1.31, 1.34)
SNF/H Non-MASH 0.22 (0.21, 0.22)
MASH 0.18 (0.18, 0.18)
1.20 (1.17, 1.23)
Prescriptions Non-MASH 29.66 (29.59, 29.72)
MASH 31.81 (31.61, 32.02)
1.07 (1.07, 1.08)
0.5 1.0 2.0
All-Cause Costs (Mean, USD PPPY)
Non-MASH
MASH
IP $4,604 $6,855
OP $9,565 $11,107
ER $498 $635
SNF/H $1,653 $1,874
Total medical $16,503 $20,256
Prescriptions $4,039 $4,634
Total $20,542 $24,890
$0 $10,000 $20,000 $30,000
Difference (95% CI)
$2,251 ($2,130, $2,372)
$1,542 ($1,406, $1,679)
$137 ($128, $146)
$221 ($166, $276)
$3,753 ($3,516, $3,990)
$595 ($471, $719)
$4,348 ($4,062, $4,634)
Ratio (95% CI)
1.49 (1.46, 1.52)
1.16 (1.15, 1.18)
1.28 (1.26, 1.30)
1.13 (1.10, 1.17)
1.23 (1.21, 1.24)
1.15 (1.12, 1.18)
1.21 (1.20, 1.23)
0.5 1.0 2.0

CI, confidence interval; ER, emergency room; IP, inpatient; IPW, inverse probability weighting; MASH, metabolic dysfunction-associated steatohepatitis; OP, outpatient; PPPY, per-patient per-year; SNF/H, skilled nursing facility and hospice; USD, US dollars.

Figure 3. HCRU and Costs comparisons between cirrhotic MASH and non-cirrhotic MASH cohorts after IPW.

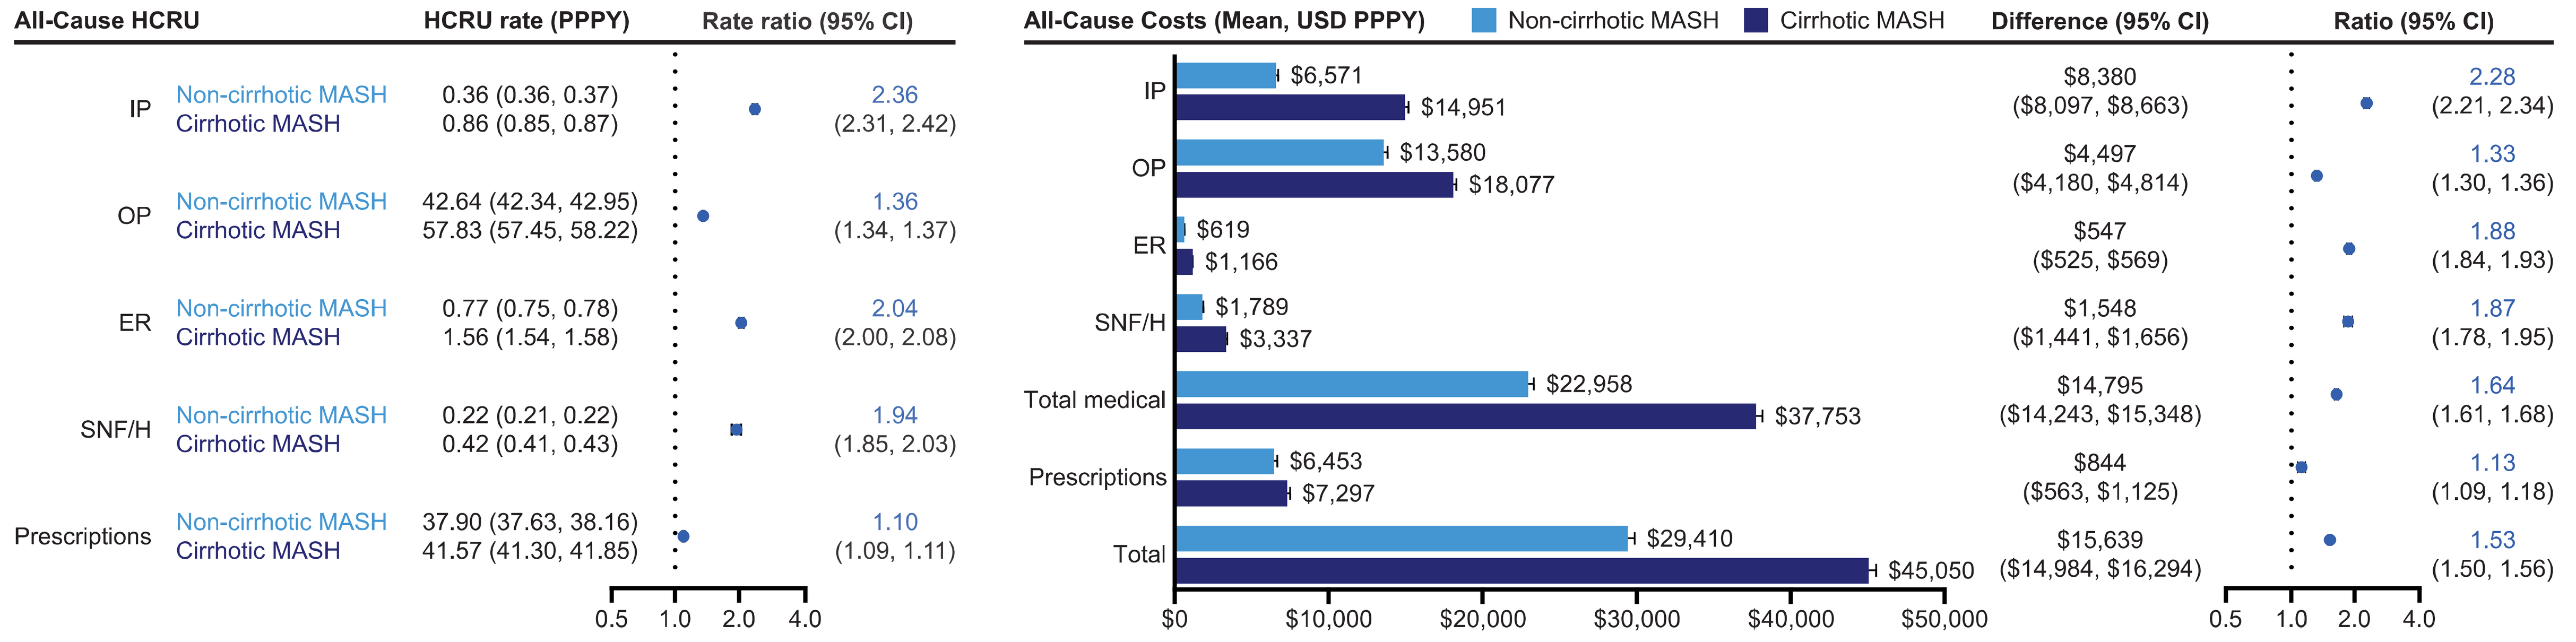
All-Cause HCRU
HCRU rate (PPPY)
Rate ratio (95% CI)
IP Non-cirrhotic MASH 0.36 (0.36, 0.37)
Cirrhotic MASH 0.86 (0.85, 0.87)
2.36 (2.31, 2.42)
OP Non-cirrhotic MASH 42.64 (42.34, 42.95)
Cirrhotic MASH 57.83 (57.45, 58.22)
1.36 (1.34, 1.37)
ER Non-cirrhotic MASH 0.77 (0.75, 0.78)
Cirrhotic MASH 1.56 (1.54, 1.58)
2.04 (2.00, 2.08)
SNF/H Non-cirrhotic MASH 0.22 (0.21, 0.22)
Cirrhotic MASH 0.42 (0.41, 0.43)
1.94 (1.85, 2.03)
Prescriptions Non-cirrhotic MASH 37.90 (37.63, 38.16)
Cirrhotic MASH 41.57 (41.30, 41.85)
1.10 (1.09, 1.11)
0.5 1.0 2.0 4.0
All-Cause Costs (Mean, USD PPPY)
Non-cirrhotic MASH
Cirrhotic MASH
IP $6,571 $14,951
OP $13,580 $18,077
ER $619 $1,166
SNF/H $1,789 $3,337
Total medical $22,958 $37,753
Prescriptions $6,453 $7,297
Total $29,410 $45,050
$0 $10,000 $20,000 $30,000 $40,000 $50,000
Difference (95% CI)
$8,380 ($8,097, $8,663)
$4,497 ($4,180, $4,814)
$547 ($525, $569)
$1,548 ($1,441, $1,656)
$14,795 ($14,243, $15,348)
$844 ($563, $1,125)
$15,639 ($14,984, $16,294)
Ratio (95% CI)
2.28 (2.21, 2.34)
1.33 (1.30, 1.36)
1.88 (1.84, 1.93)
1.87 (1.78, 1.95)
1.64 (1.61, 1.68)
1.13 (1.09, 1.18)
1.53 (1.50, 1.56)
0.5 1.0 2.0 4.0

CI, confidence interval; ER, emergency room; IP, inpatient; IPW, inverse probability weighting; MASH, metabolic dysfunction-associated steatohepatitis; OP, outpatient; PPPY, per-patient per-year; SNF/H, skilled nursing facility and hospice; USD, US dollars.

Limitations

- MASH is often undiagnosed and therefore, HCRU and cost attributed to MASH could be underestimated.
- MASH may contribute to an increased risk of cardiovascular morbidity. In our IPW models, adjusting for cardiovascular disease could represent overadjustment for outcomes associated with MASH; however, this conservative approach was adopted and may have resulted in an underestimation of HCRU and costs attributed to MASH.
- Our study population may not be representative of all patients with MASH, as it only included individuals aged 66 and older with Medicare FFS plans.

Conclusion

- Among Medicare beneficiaries ≥66 years old, MASH was associated with significantly greater HCRU and costs, even after adjustments for cardiometabolic and other comorbidities.
- Higher HCRU and costs for patients with MASH is likely driven by the management of liver disease, which may include advanced liver fibrosis, cirrhosis, and hepatic decompensation.
- HCRU and costs, adjusted for baseline characteristics, were also greater for patients with cirrhotic MASH compared with beneficiaries with non-cirrhotic MASH.
- Understanding the real-world healthcare burden of MASH is important for guiding healthcare decisions including screening, diagnosis, staging and treatment.

This study was sponsored by Novo Nordisk. The authors thank Kristen Downs, Justine Mailliot and Yingjie Ding of Genesis Research Group, Hoboken, NJ, US, who provided medical writing and editorial support.

Presented at the AMCP Nexus 2025 conference, October 27–30, National Harbor, MD, USA.

Disclosures:
HA, AA, AH, and CZ are employees of Novo Nordisk Inc. and are shareholders of Novo Nordisk A/S.
AT, JV, and JH are employees of Genesis Research Group, which received consulting fees from Novo Nordisk.
PM is a consultant/advisor to Novo Nordisk Inc.

References:
(1) Lekakis V, et al. *Eur J Intern Med*. Apr 2024;122:3–10. **(2)** Younossi ZM, et al. *Hepatol Commun*. 2024;8(8); **(3)** Kim Y, et al. *J Comp Eff Res*. 2024;13(12):e240096; **(4)** Gordon SC, et al. *Am J Gastroenterol*. 2020;115(4):562–574.; **(5)** Tapper EB, et al. *J Med Econ*. 2023;26(1):348–356; **(6)** Gordon SC, et al. *Hepatol Commun*. 2020;4(7):998–1011.